Development of a new bronchodilator to enable better treatment of Asthma, COPD, and CF

Francis Green[1,2], John Dennis[1], Richard Leigh[2], Grishma Shrestha[1], Sharif Galal[1], Morenike Fadayomi[1], Mustafa Al-Saiedy[2], Andrea Chiu[1], Cora Pieron[1], Tamer El Mays[1], David Nelson[1], Veronica Swystun[2], Gareth Lewis[1]

[1]SolAeroMed Inc, 3535 Research Rd, Calgary NW T2L 2K8, Canada

[2]Airway Inflammation Group, University of Calgary and Alberta Health Services, Calgary, Canada

Abstract

- A new drug (**S-1226**) to treat asthma incorporating a novel formulation combining the potent bronchodilator carbon dioxide (CO_2) delivered to the lung as a gas in combination with a synthetic lung surfactant, perfluoroctyl bromide (PFOB) co-delivered as an aerosol
- **S-1226** is being developed by academic co-founders through the Canadian based biotech start-up SolAeroMed Inc.
- **S-1226**:
 - Effective: treats methacholine and allergen induced bronchoconstriction in sheep and rat models of asthma
 - Safe: Phase I clinical trial showed minimal adverse effects at all dosing levels (4%, 8%, 12% CO_2)
 - Currently in Phase II clinical trial – allergen induced asthmatic subjects
 - Mechanism of action:
 - A relaxant effect of CO_2 on constricted airway smooth muscle
 - The ability of the gaseous component to rapidly penetrate closed airways
 - The ability to lubricate mucous plugs thus enhancing mucous clearance
- **S-1226** and salbutamol do not compete for the same receptors
 - Aims to complement rather than replace current treatments and is likely to be used when conventional bronchodilators are ineffective in opening closed airways
- A novel portable delivery device is currently under development
- Anticipated future roles for **S-1226**:
 - Treating exacerbations of COPD and CF
 - A combination therapy to enhance delivery and effectiveness of other respiratory drugs

Medical Need for a New Asthma Drug

- Asthma:
 - Affects 300 million individuals worldwide; 400 million expected by 2025
 - Worldwide 250,000 asthma deaths every year
- **$8.9 billion** problem in asthma when conventional emergency treatments don't work
- Out of **3.9 million** emergency hospital visits per year worldwide:
 - 85% sent home after 3 hours (**$2.1 billion)**
 - 15% use 1.8 million hospital bed-days (**$6.8 billion)**
- Short-acting β_2-agonists are the most common front-line emergency treatment
 - Only one short acting bronchodilator (salbutamol)
 - One-fifth to one-third of patients do **not** respond well
 - Tachyphylaxis and toxicity have been demonstrated
- Major challenge for treating acute asthma is to open occluded airways rapidly enough to re-establish ventilation and allow delivery of O_2 and conventional medication to airways

Background to the New Treatment: S-1226™

- In severe asthma the airways are blocked by a combination of bronchoconstriction and mucous plugs rendering personal inhalers ineffective
- A new type of treatment is required that has both bronchodilator and biophysical properties (mucous plug penetration and clearance)
- This led to the discovery of **S-1226**, a new class of short-acting bronchodilators by University of Calgary scientists
- SolAeroMed Inc. was formed to progress R&D and support commercial development of **S-1226**
- Patents have been issued in the USA and Europe

Novel Asthma Drug S-1226™

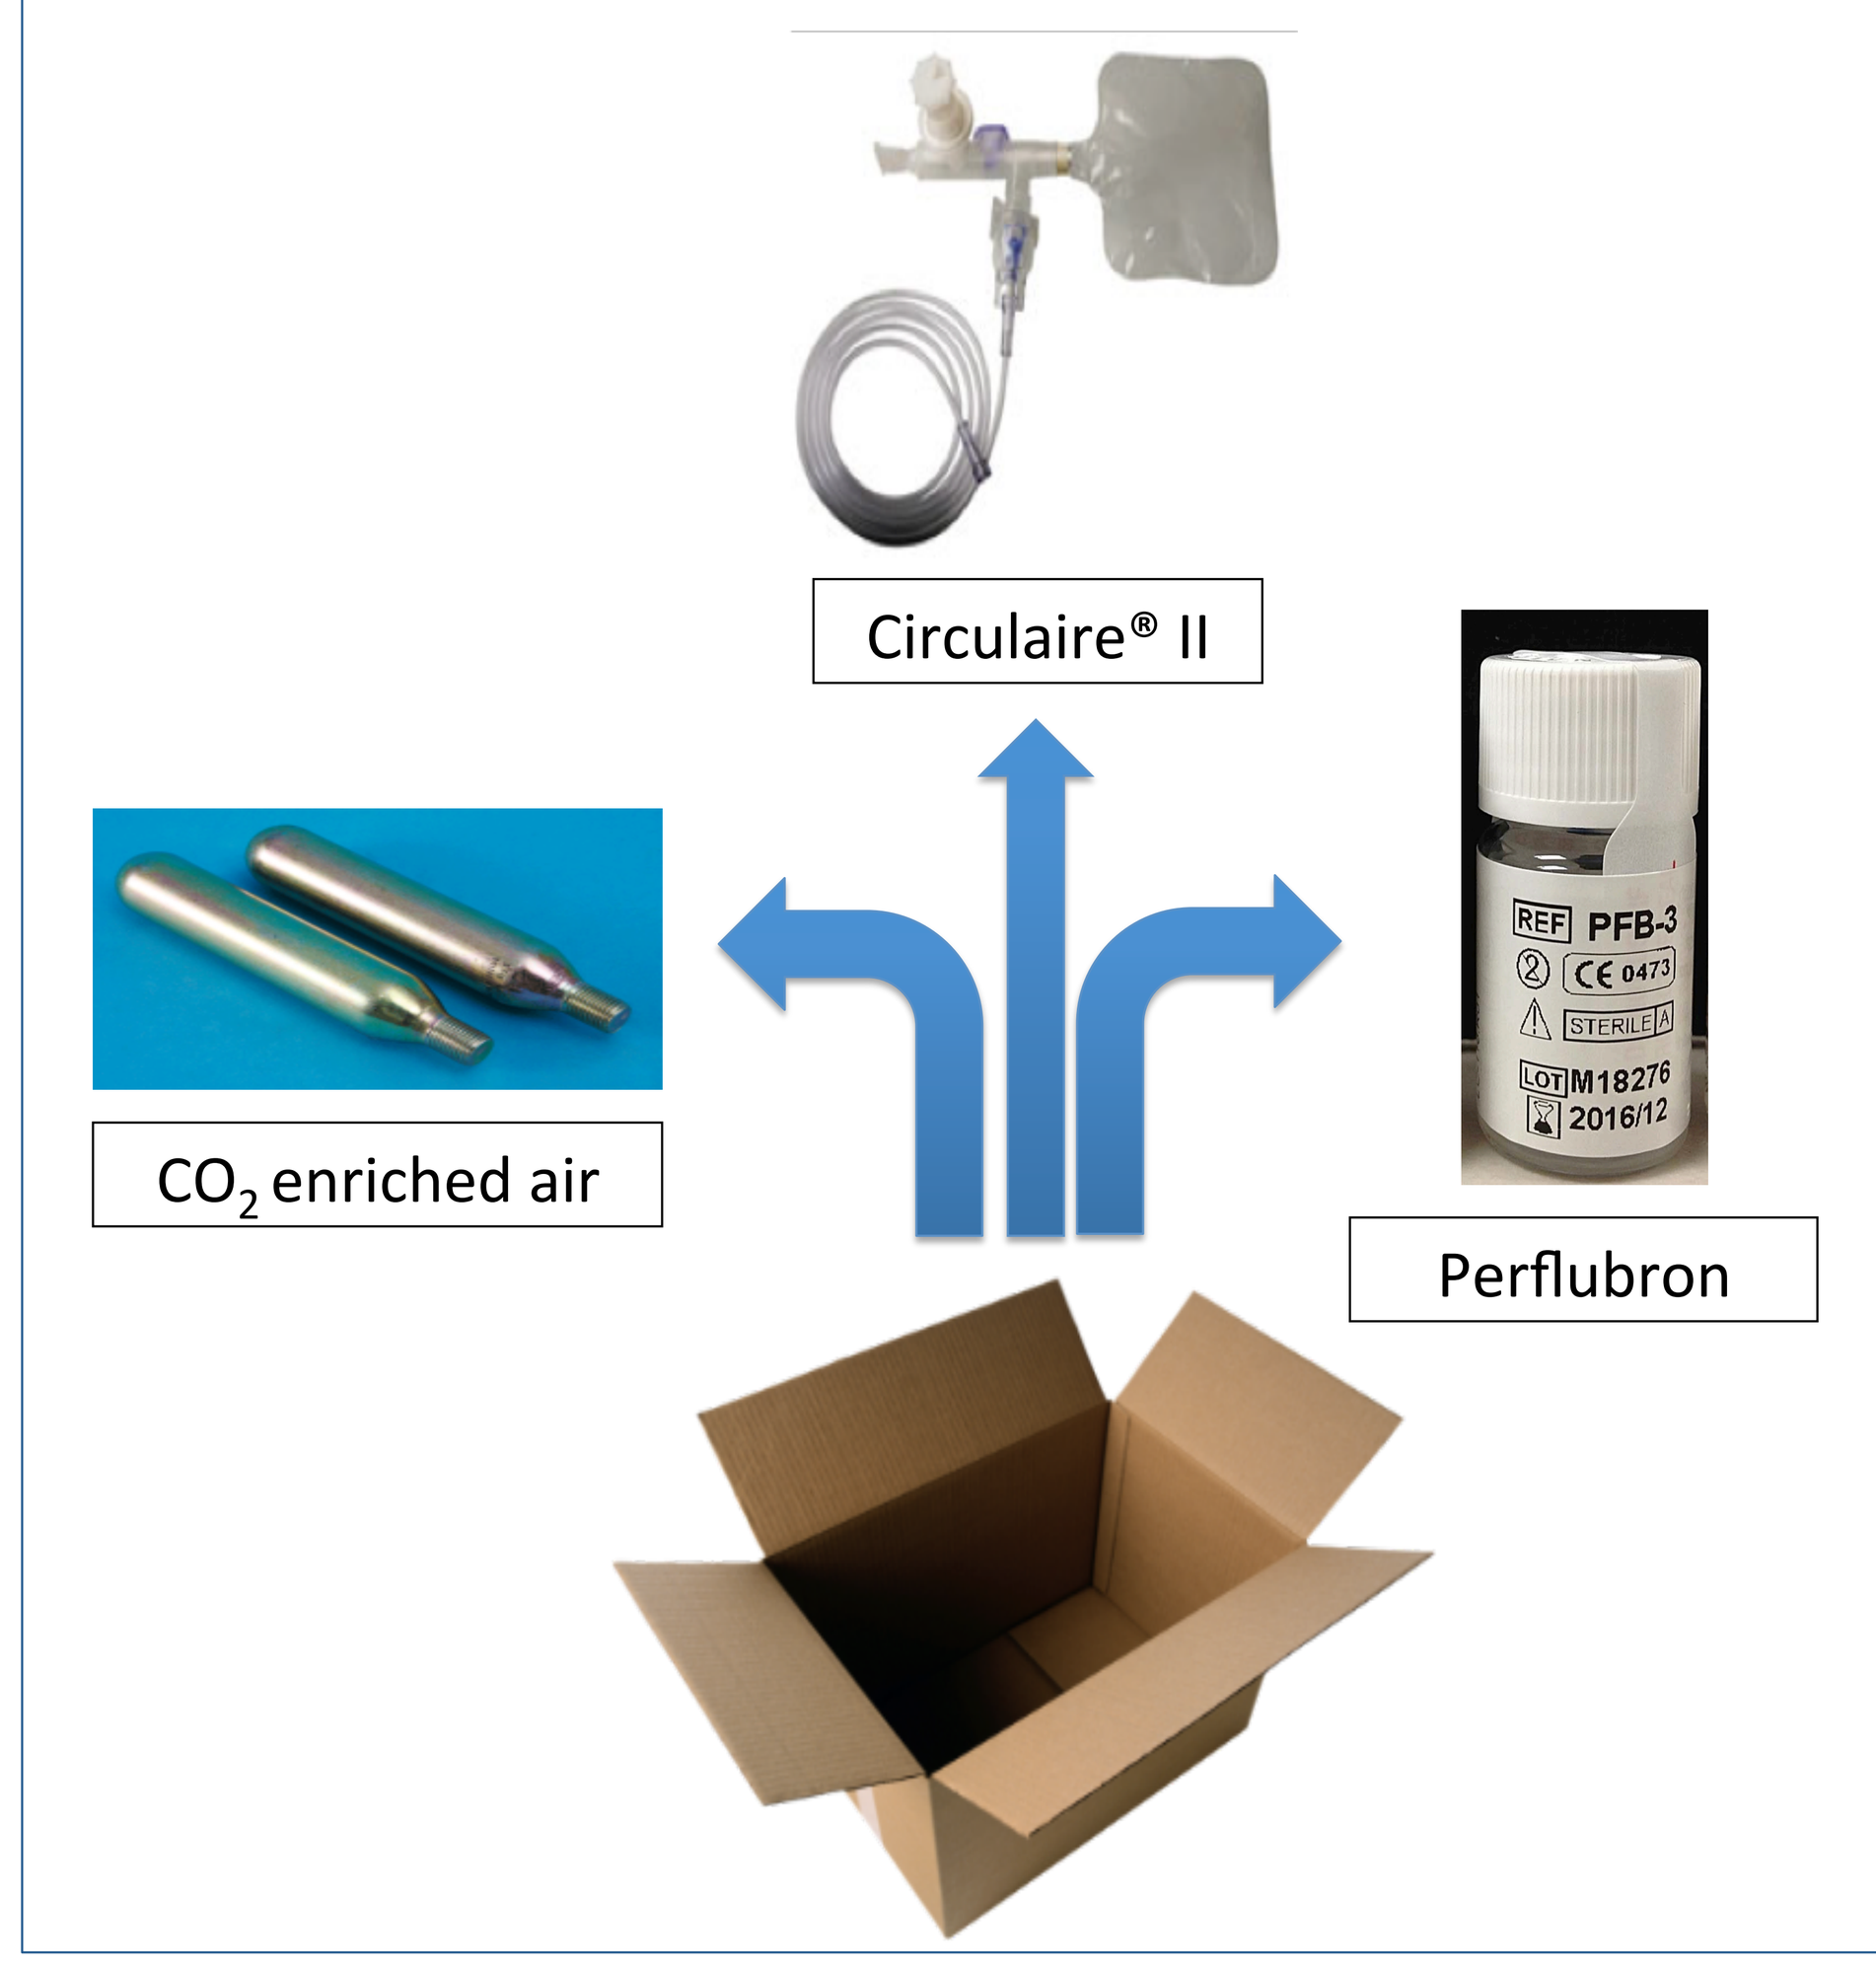
Circulaire® II
CO_2 enriched air
Perflubron
REF PFB-3
LOT M18276
2016/12

- **S-1226** consists of 3mL Perflubron synthetic surfactant nebulized with CO_2-enriched air (4-12% CO_2, 20% O_2, with balanced N_2)
 - Delivered as an aerosol/vapour/gas mixture
 - New mechanism of action
 - Both CO_2 and Perflubron are well-described and safe
 - Both CO_2 and Perflubron have previously been approved by FDA, EMA, HC

Pre-Clinical Studies

Sheep Model of Asthma

Figure 1

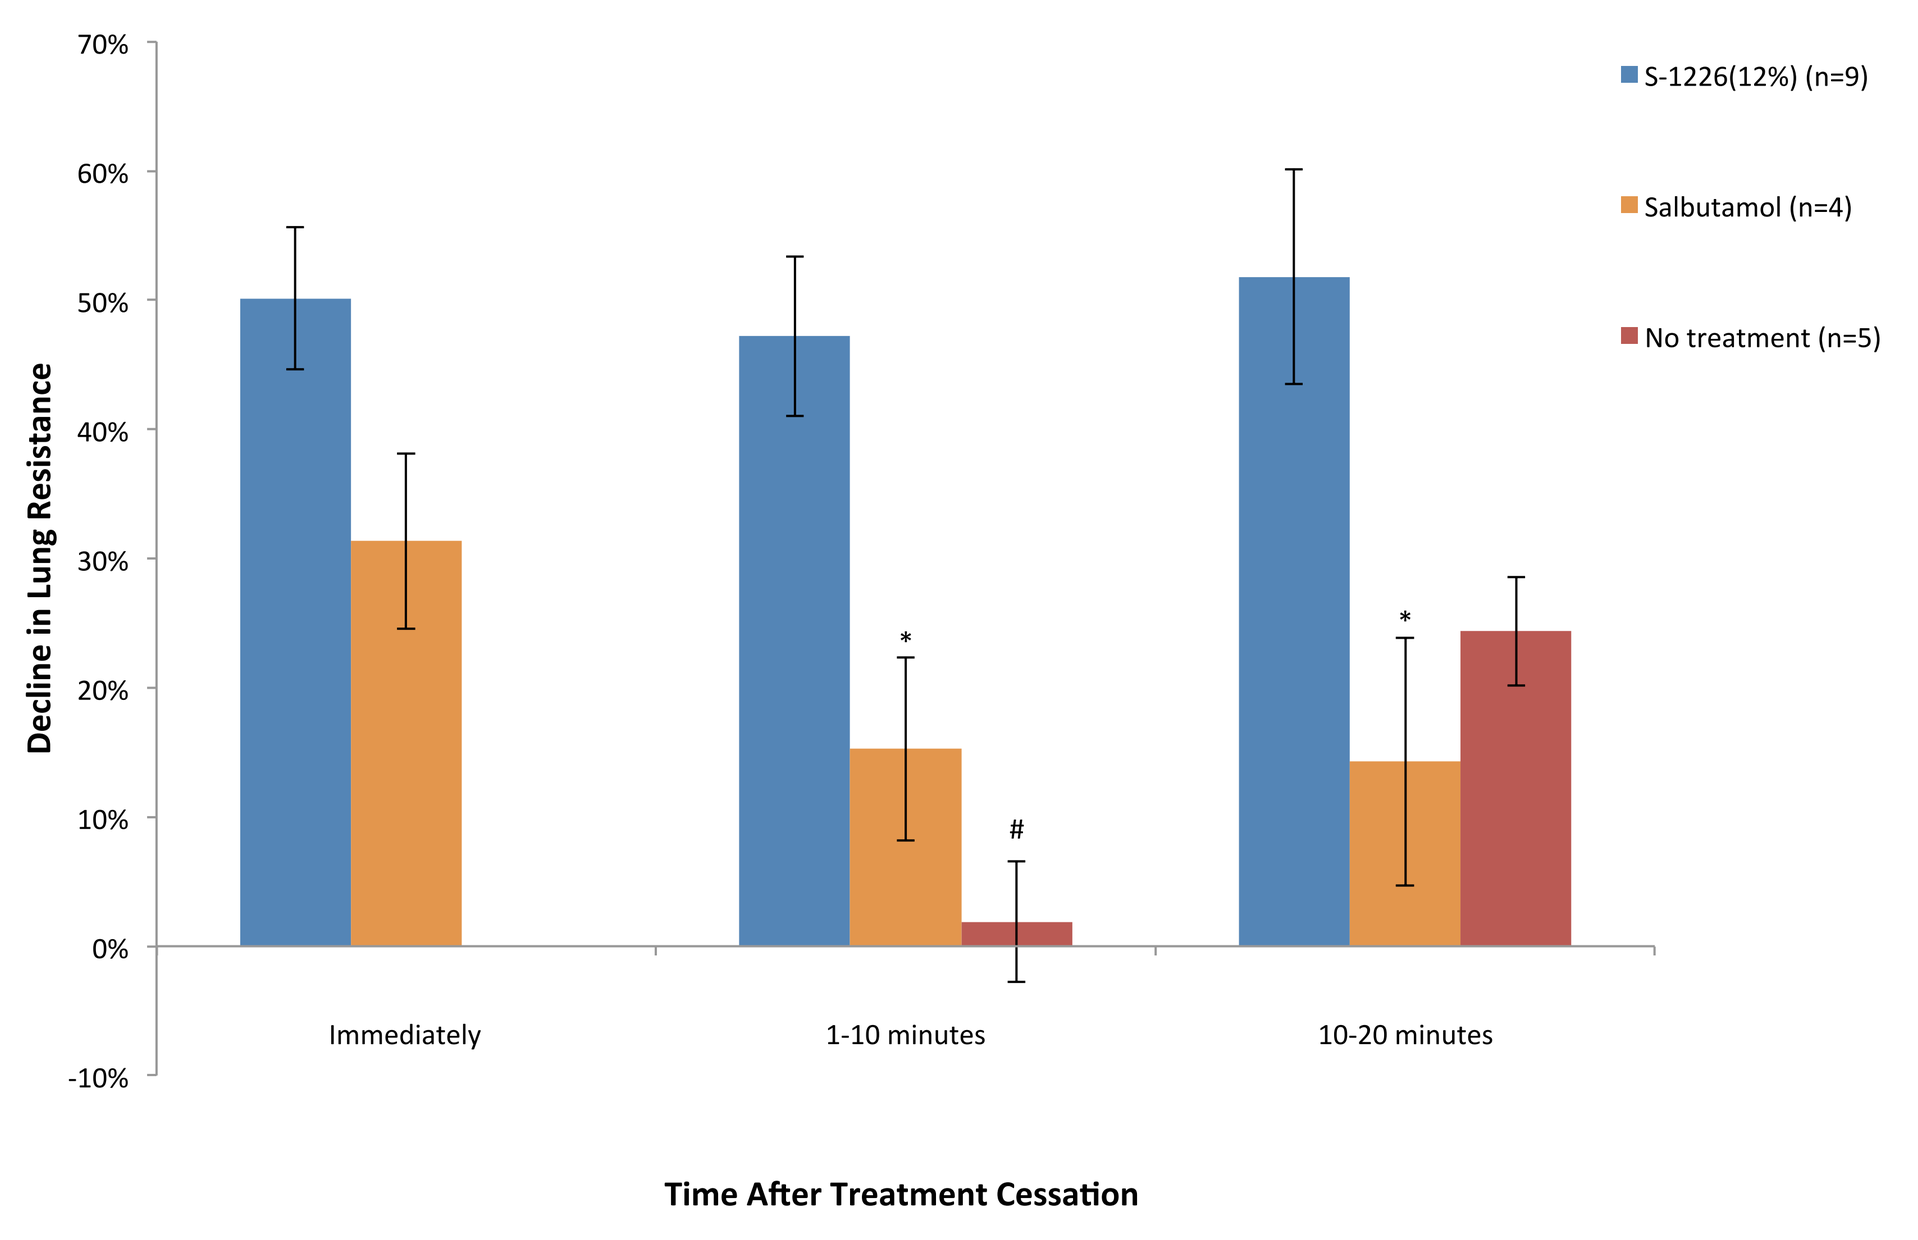
70%
60%
50%
40%
30%
20%
10%
0%
-10%
Decline in Lung Resistance
S-1226(12%) (n=9)
Salbutamol (n=4)
No treatment (n=5)
Immediately
1-10 minutes
10-20 minutes
Time After Treatment Cessation

Figure 2

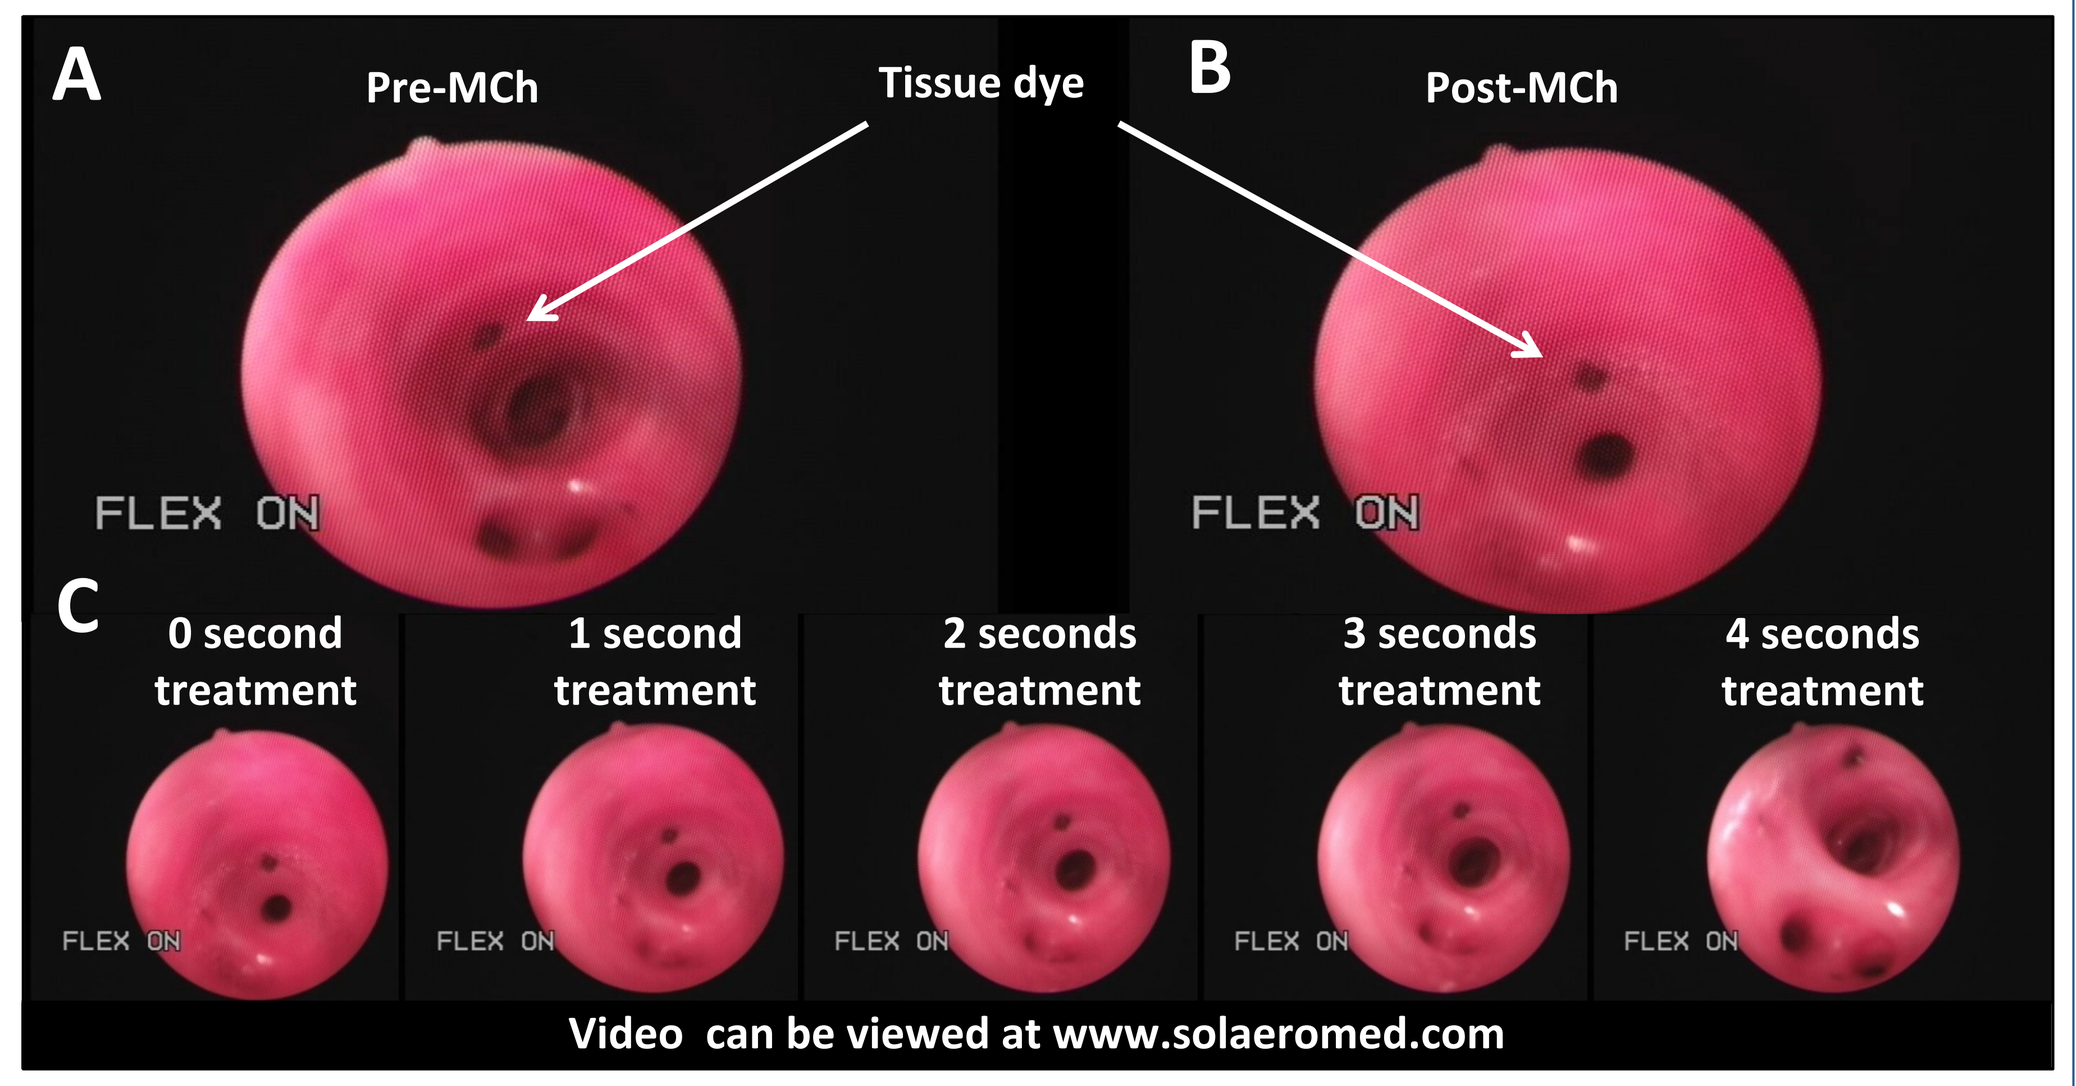
A
Pre-MCh
Tissue dye
B
Post-MCh
C
0 second treatment
1 second treatment
2 seconds treatment
3 seconds treatment
4 seconds treatment
Video can be viewed at www.solaeromed.com

Figure 1. Decline in lung resistance following **S-1226** treatment in house dust mite (HDM) challenged sheep. Treatment with **S-1226** showed rapid and sustained decline in lung resistance compared to no treatment and salbutamol. P-values show significant difference from **S-1226**; *p<0.05, #p≤0.01.

Figure 2. Bronchoscopic still images of sheep airways before **(A)** and after **(B)** methacholine (MCh) challenge and seconds after initiating treatment with nebulized **S-1226 (C)**. Complete airway opening is seen 4 seconds after the start of **S-1226** treatment.

Mucolytic (*In Vitro)*

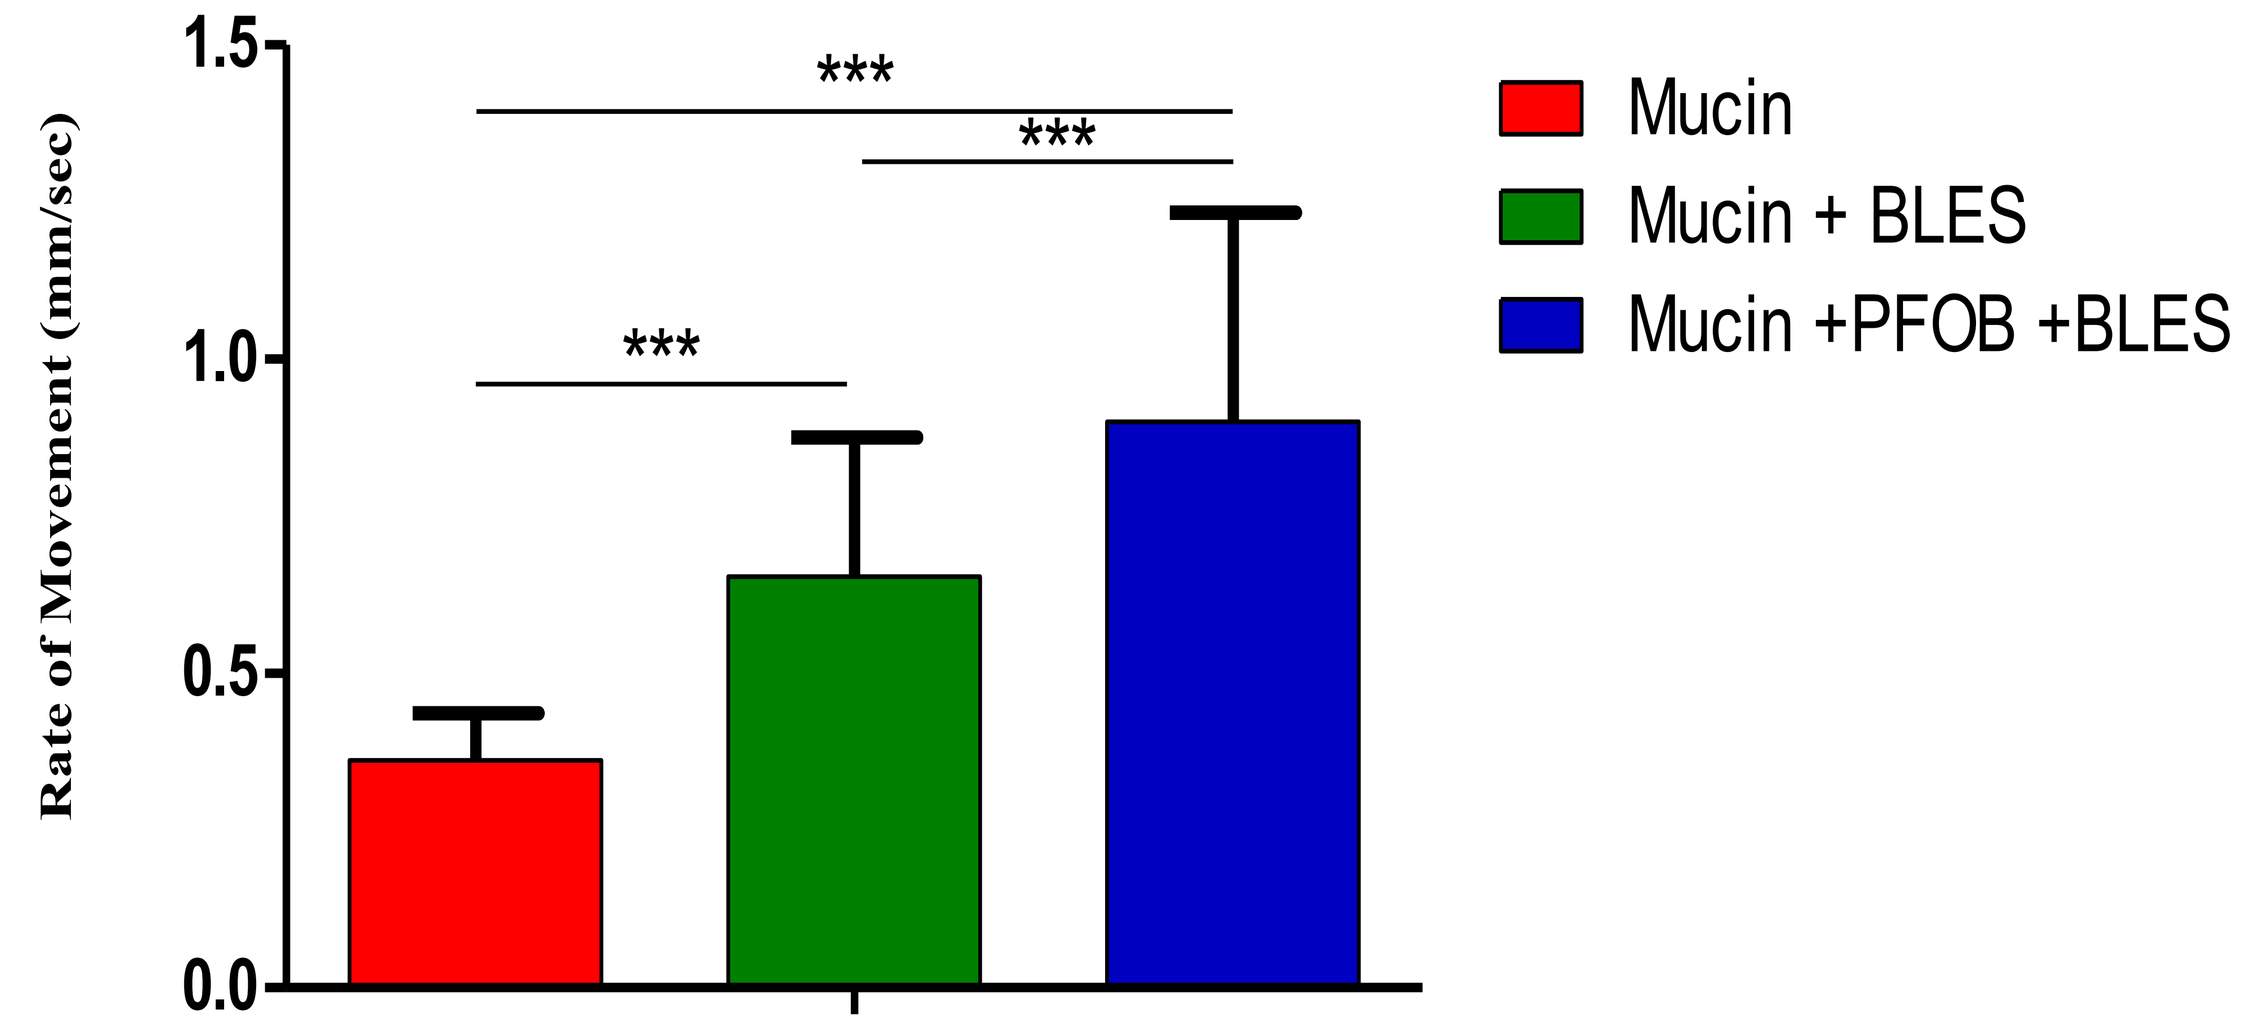
1.5
1.0
0.5
0.0
Rate of Movement (mm/sec)

Mucin
Mucin + BLES
Mucin +PFOB +BLES

Figure 3. Clearance (mm/sec) of mucin is enhanced in the presence of pulmonary surfactant (BLES) and further enhanced in the presence of Perflubron (PFOB). ***p<0.01

Clinical Studies

	Phase I S-1226 at 4%, 8%, and 12% CO_2	Phase IIa S-1226 at 8% CO_2
Completion	Q2 2014	Expected: Q3 2015
Population	Healthy individuals	Mild atopic asthmatics
Adverse Events	Mild	Expected: minimal based on Phase I
Drug Administration and Characterization	• WestMed Circulaire® II Hybrid nebulizer system • Perflubron nebulized with compressed gas (flow rate: ~8.9 L/min, driving pressure: 20 psi) • Perflubron delivered at a mean ratio of 41:59 (aerosol:vapour) • Aerosol droplets have a 2.4 µm mass median aerodynamic diameter with 2.9 geometric standard deviation	

Summary and Future Directions

- **S-1226**:
 - **Rapidly dilated airways (*in* vivo)**
 - **Enhances mucous plug clearance (*in* vitro)**
 - **New mechanism of action**
 - **First-in-class product; pharmacological and biophysical**
 - **Platform technology for delivery of other drugs**

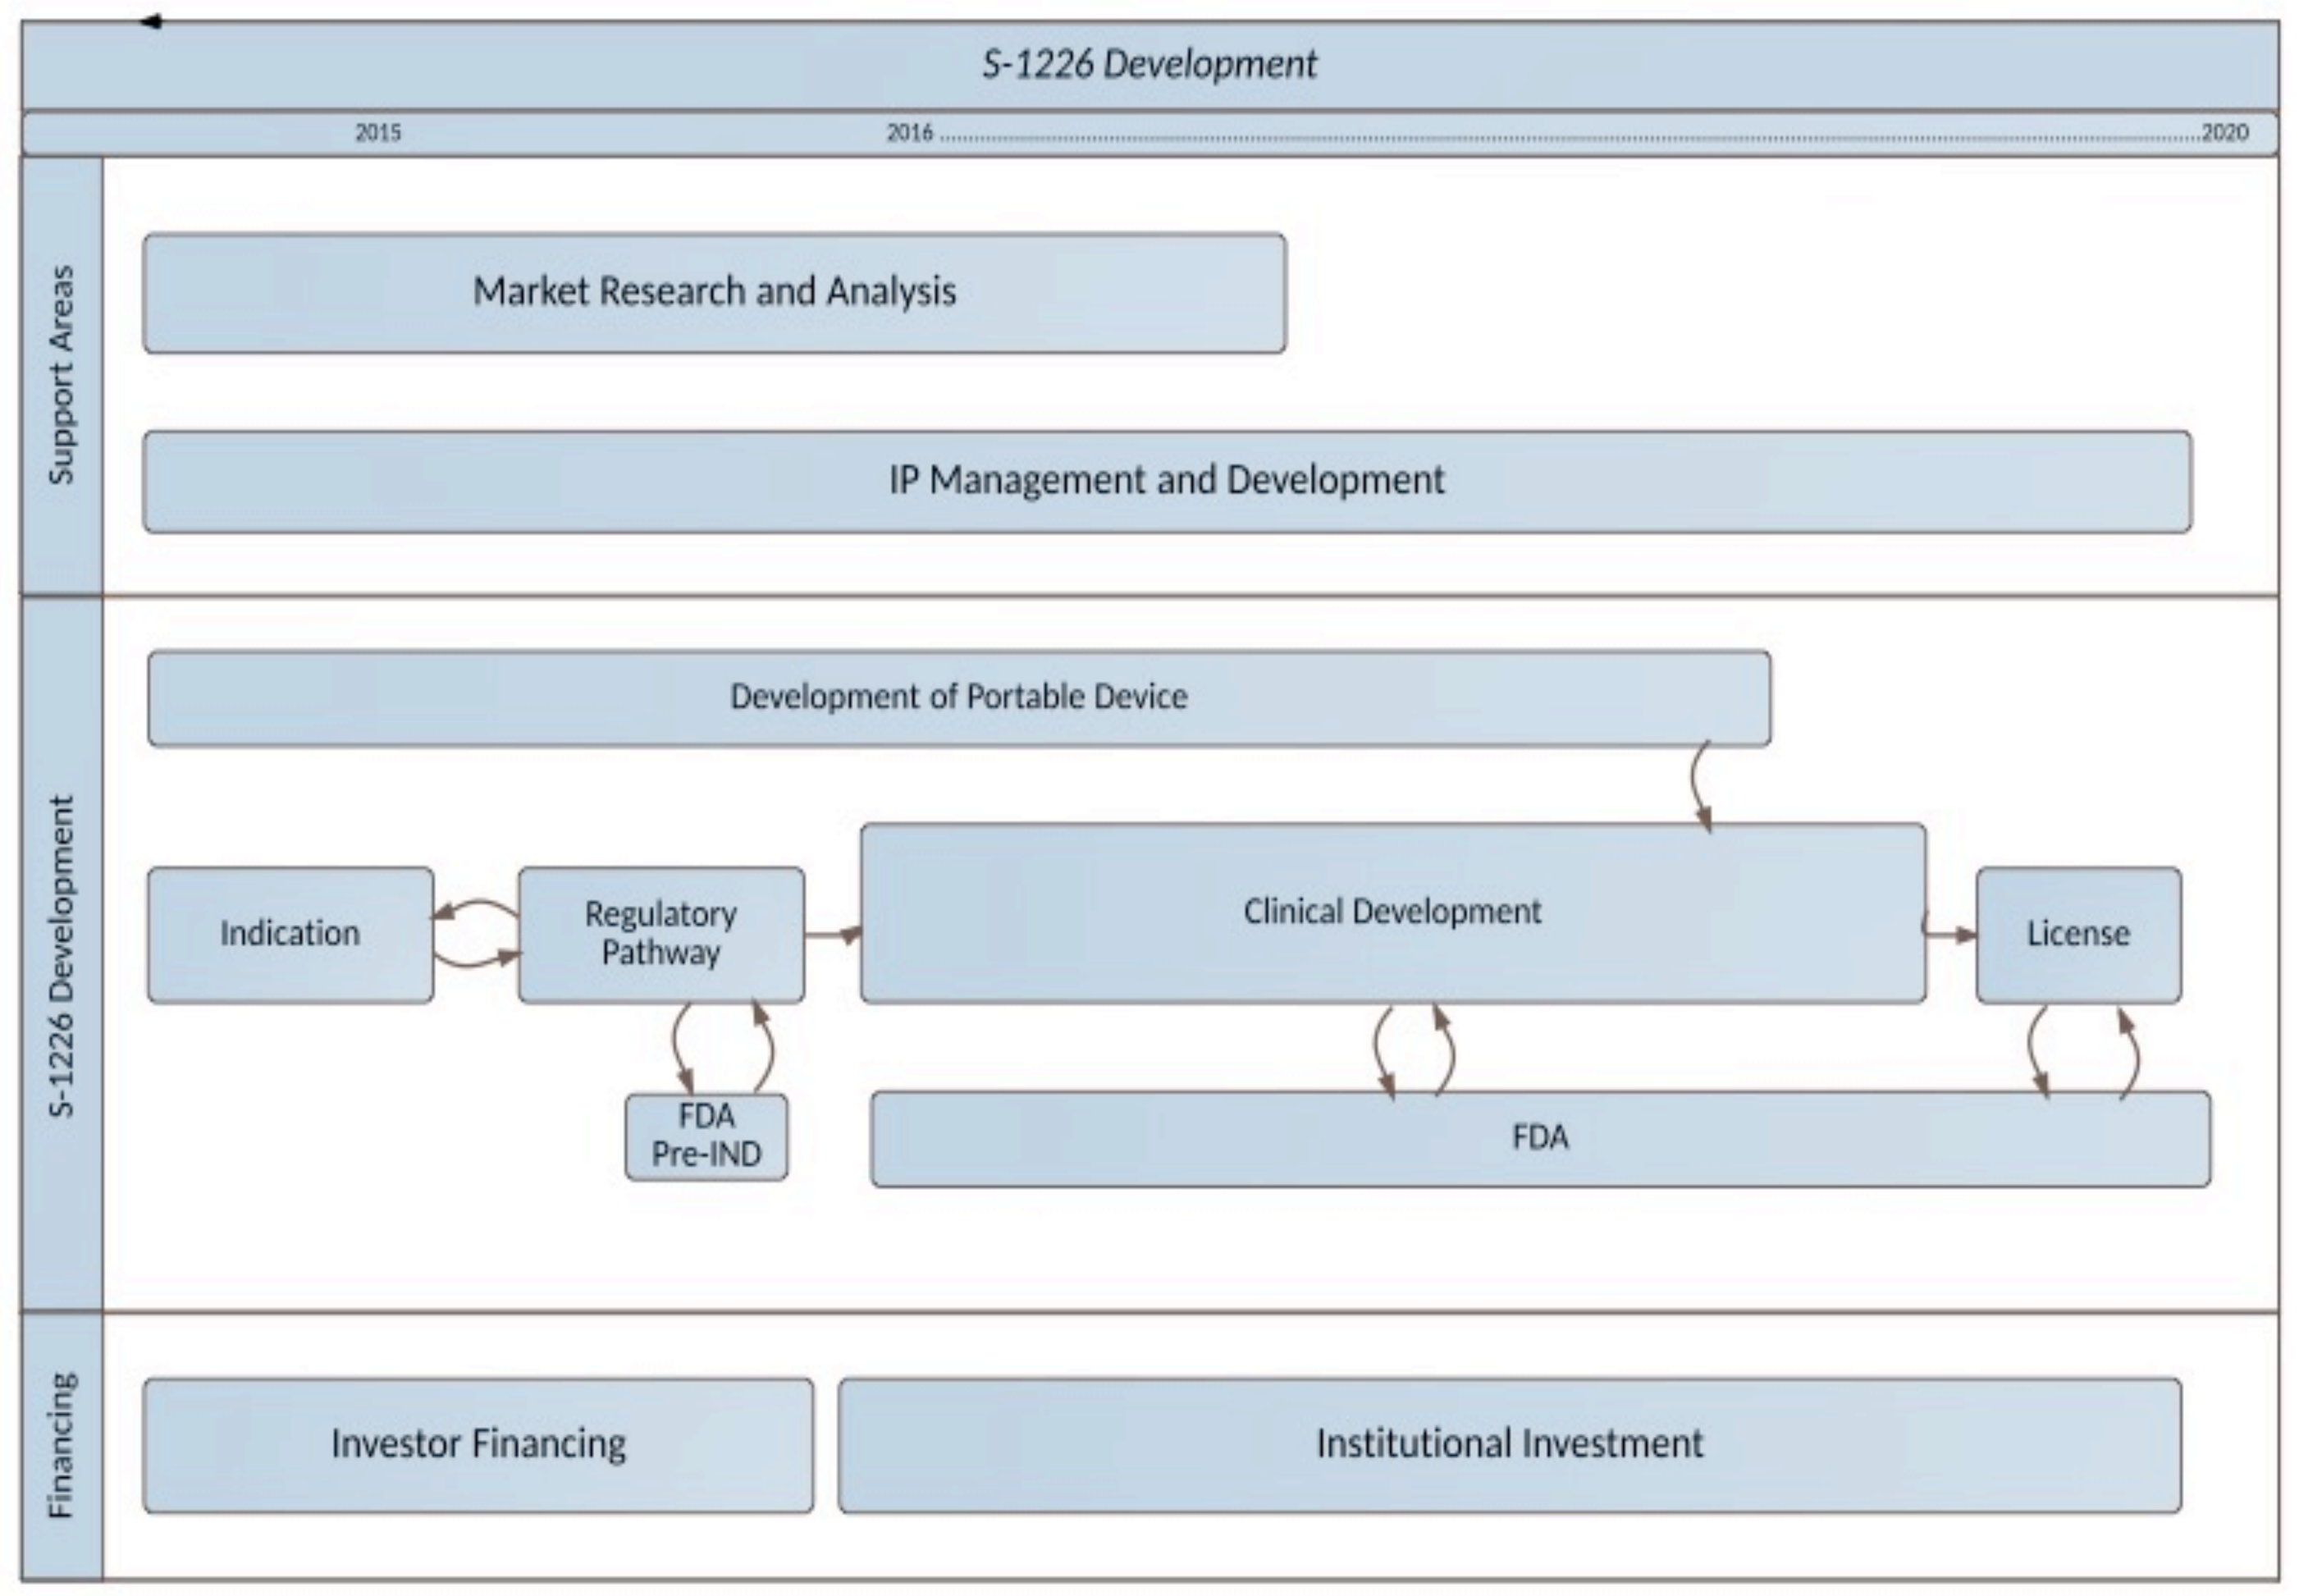
S-1226 Development
2015
2016
2020
Support Areas
Market Research and Analysis
IP Management and Development
S-1226 Development
Development of Portable Device
Indication
Regulatory Pathway
Clinical Development
License
FDA Pre-IND
FDA
Financing
Investor Financing
Institutional Investment

- Development of a personal rescue device to be used by patients as an emergency technique when away from the hospital setting

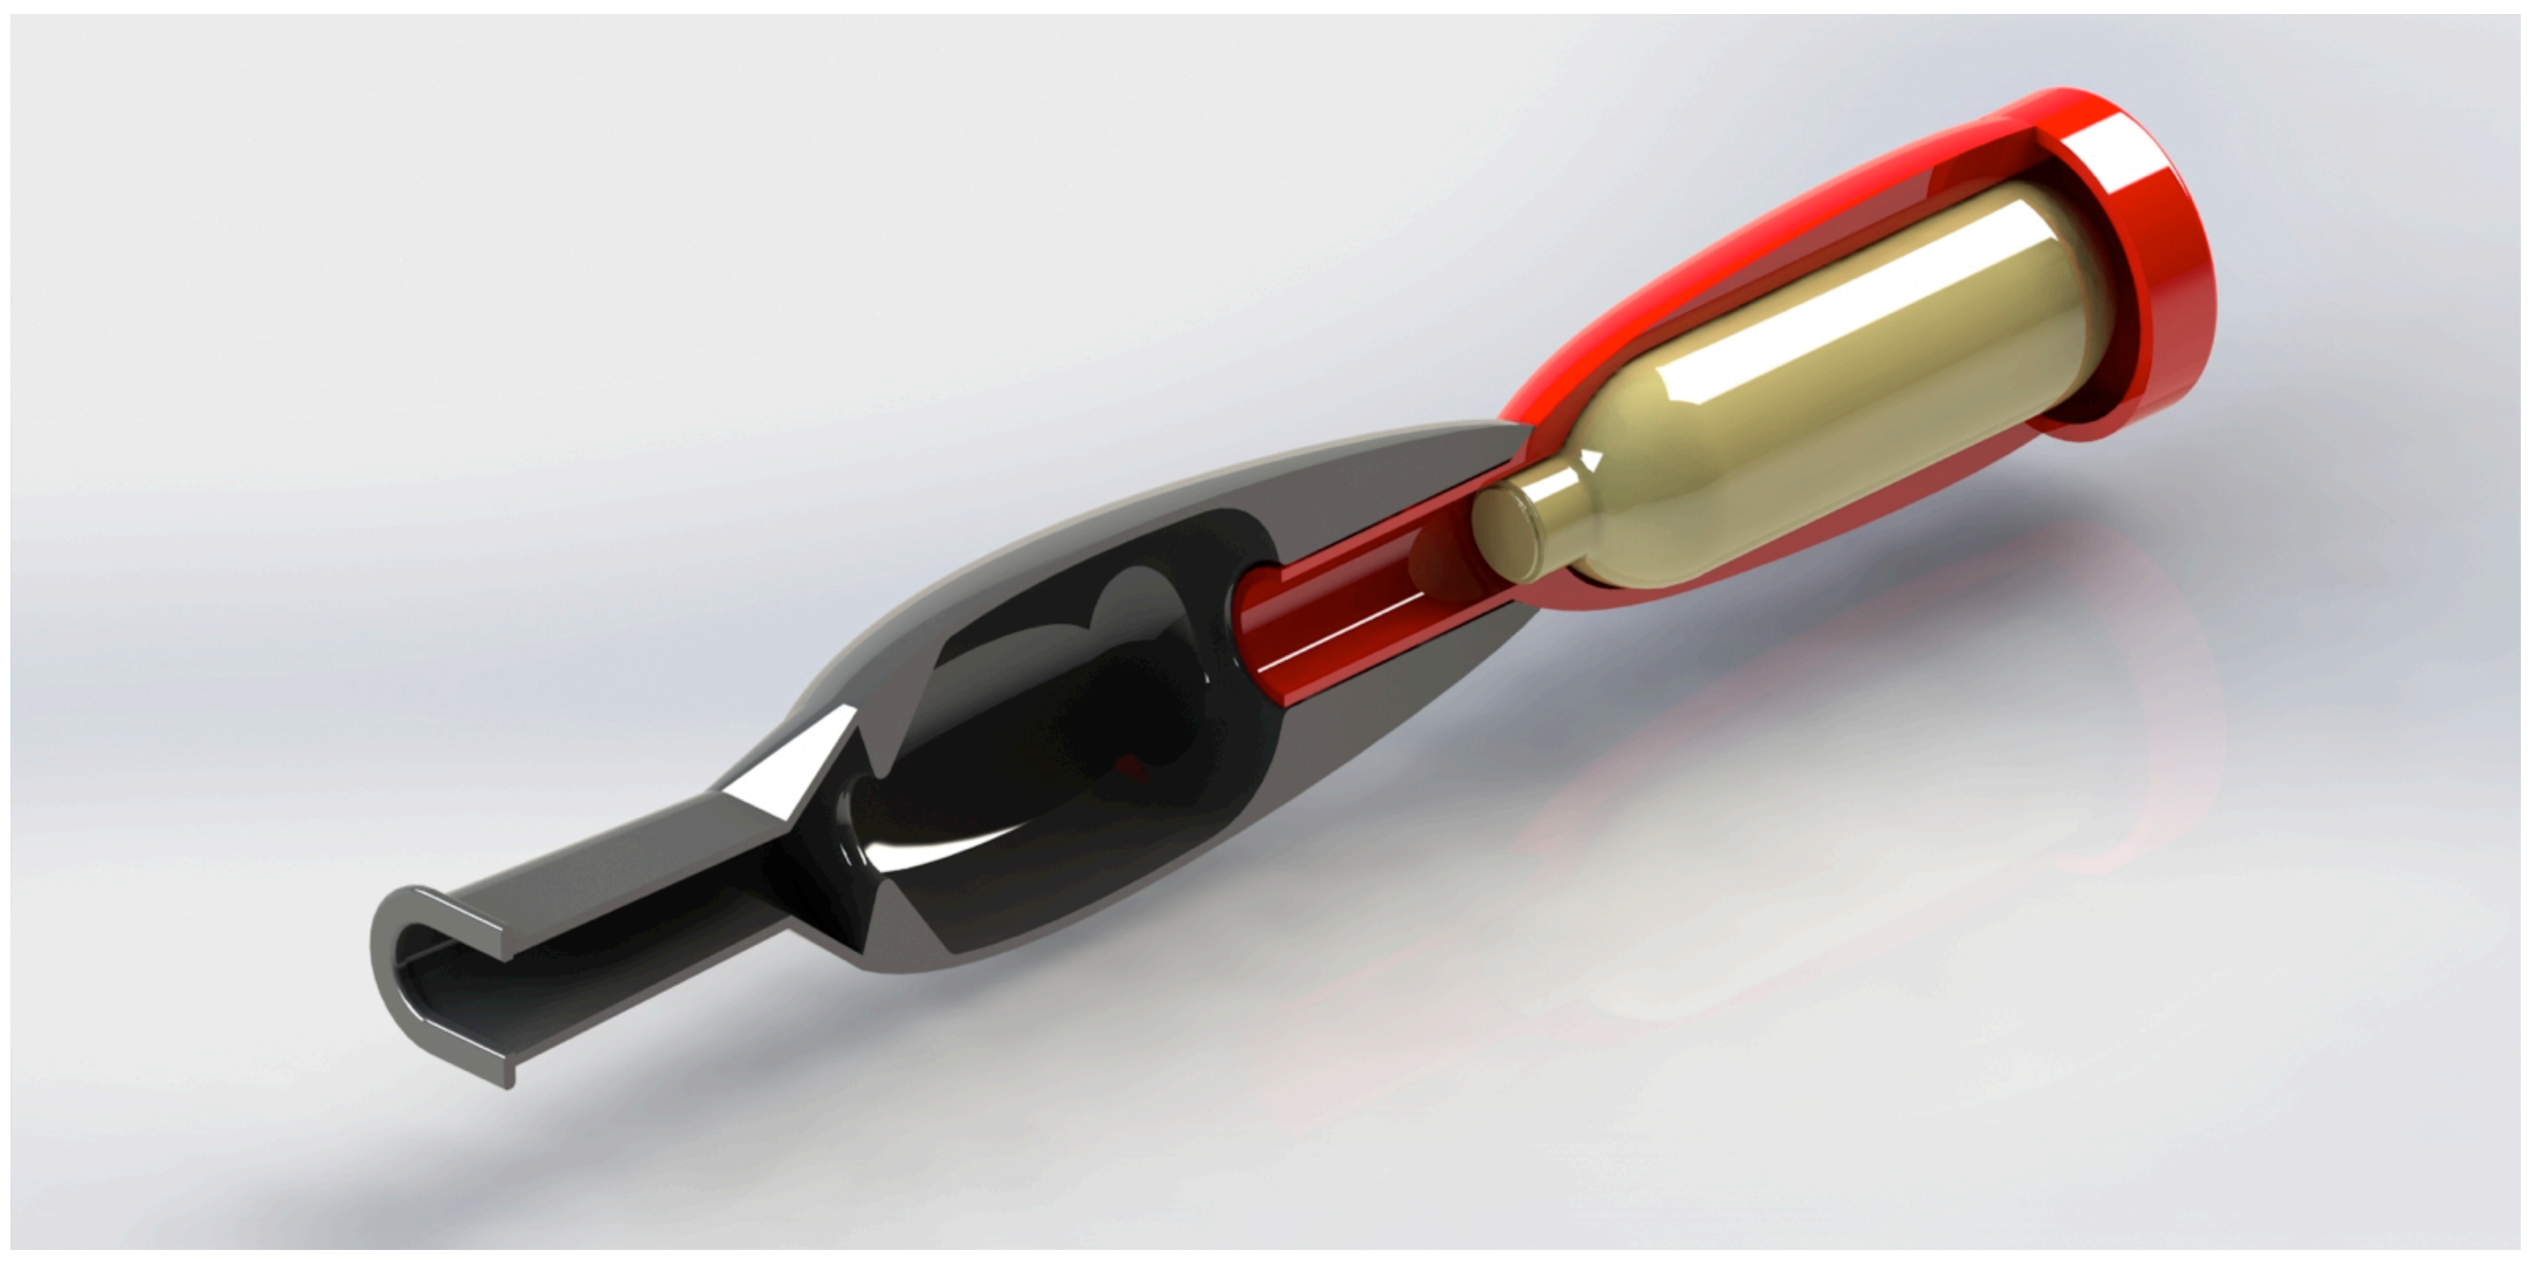