First in Class, Respiratory Drug Gas & Drug Vapor Delivered in a Revolutionary and New Portable Device*

John Dennis[1], Andy Clark[1], Ian McAffer[1], Stefan Dalberg[2], Cora Pieron[1], Gareth Lewis[1], Gurkeet Lalli[1], Andrea Chiu[1], Morenike Fadayomi[1], Francis Green[1]

[1]SolAeroMed Inc, Calgary, Alberta, Canada, T2L 2K8

[2]Device Innovation, Southern Alberta Institute of Technology, Calgary, Alberta, Canada, T2M 0L4

Abstract

- **S-1226TM** is a new bronchodilator to treat asthma via a novel mechanism of action by combining the potent bronchodilator carbon dioxide (CO_2) with a synthetic lung surfactant, perfluoroctylbromide (PFOB) delivered as a nebulized mixture of vapour and aerosol.
- **S-1226TM**:
 - Effective: Treats methacholine and allergen induced bronchoconstriction in sheep and rat models of asthma
 - Safe: Phase I clinical trial showed minimal adverse effects at all dosing levels (4%, 8%, 12% CO_2)
 - Currently in Phase IIa clinical trial – allergen induced asthmatic subjects
 - Mechanism of action:
 - Relaxant effect of CO_2 on constricted airway smooth muscle
 - The ability of the gaseous component to rapidly penetrate closed airways
 - The ability to lubricate mucous plugs to enhance mucous clearance
- Anticipated future roles for S-1226TM:
 - Treat exacerbations of COPD and CF
 - Combination therapy to enhance delivery and efficacy of other respiratory drugs
- A novel portable delivery device to administer S-1226TM is under development
 - To be used as a personal rescue therapy following an acute respiratory attack
- First time a therapeutic drug gas and vapor is combined in a revolutionary portable delivery device to treat respiratory disease
- **S-1226TM** is being developed by academic co-founders through the Canadian based biotech start-up SolAeroMed Inc.

Medical Need for a New Asthma Therapy

- Asthma:
 - Affects 300 million individuals worldwide; 400 million expected by 2025
 - 250,000 asthma deaths every year worldwide
- For **3.7 million** asthma-related emergency hospital visits per year worldwide:
 - Short-acting β_2-agonists are the front-line emergency treatment
 - Only one short acting bronchodilator is used (salbutamol)
 - One-fifth to one-third of patients do **not** respond to this therapy
 - Tachyphylaxis and toxicity have been demonstrated
- In severe asthma, airways are blocked by bronchoconstriction and mucous plugs rendering personal inhalers ineffective
- Major challenge for treating acute asthma is to open occluded airways rapidly enough to re-establish ventilation and allow delivery of O_2 and conventional medication to airways

S-1226™: Novel Bronchodilator

- **S-1226TM** consists of 3mL PFOB synthetic surfactant nebulized with CO_2-enriched air (4-12% CO_2, 20% O_2, with balanced N_2)
 - Delivered as an aerosol/vapor/gas mixture with a unique biophysical mechanism of action
 - Both CO_2 and PFOB are well-described, safe and approved by FDA, EMA, HC
 - Rapid dilation of airways in sheep and rat models of asthma and enhancement of mucous plug clearance *in vitro*
 - Safety demonstrated in Phase I clinical trial at all dosing levels (4%, 8%, 12% CO_2) with Phase II Proof of Concept clinical trial ongoing

Acute Care Therapy: Multiple Doses

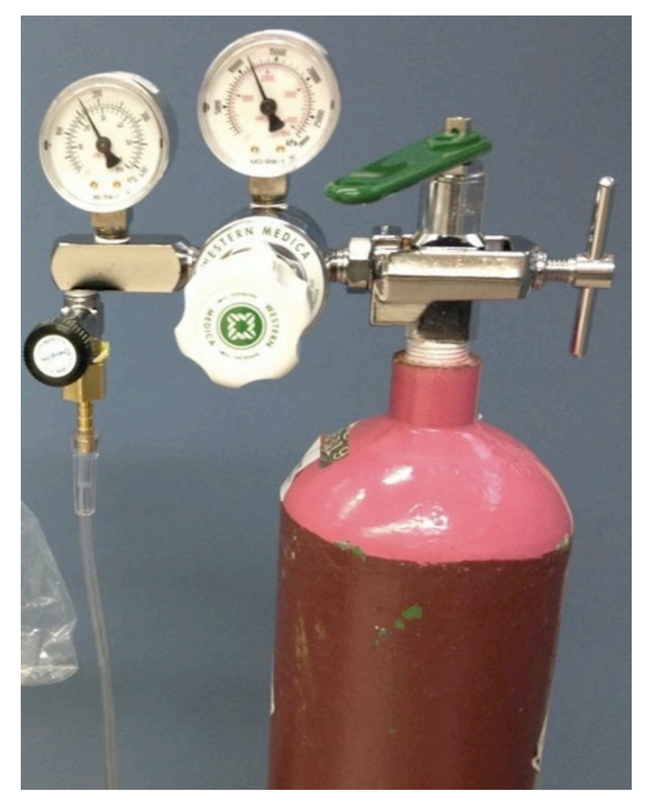

Regulator for CO_2 enriched air

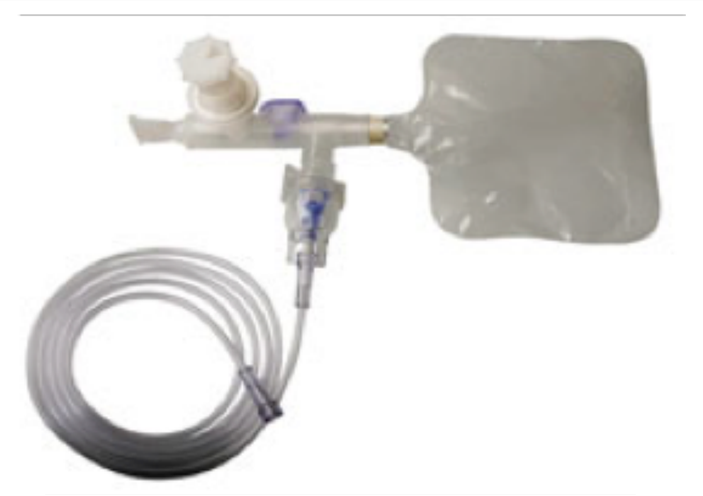

Circulaire® II Nebulizer

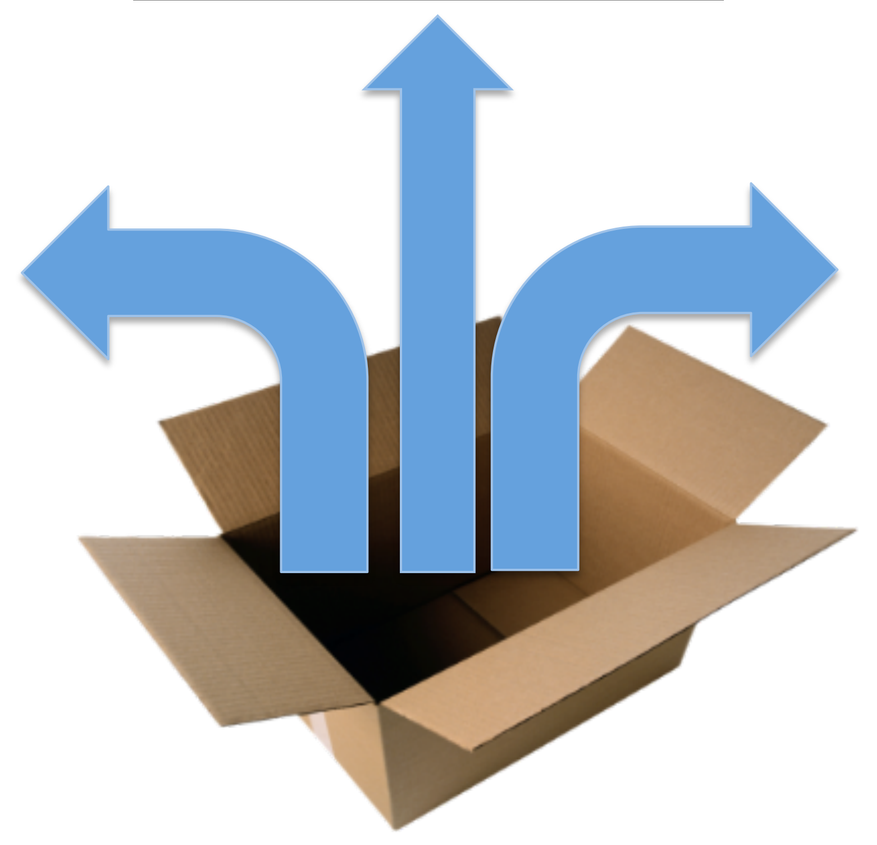

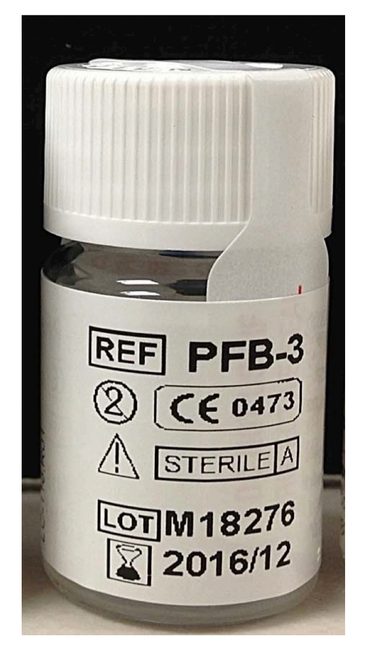
REF PFB-3
CE 0473
STERILE A
LOT M18276
2016/12

PFOB

Outpatient Rescue Device: Disposable Single Use

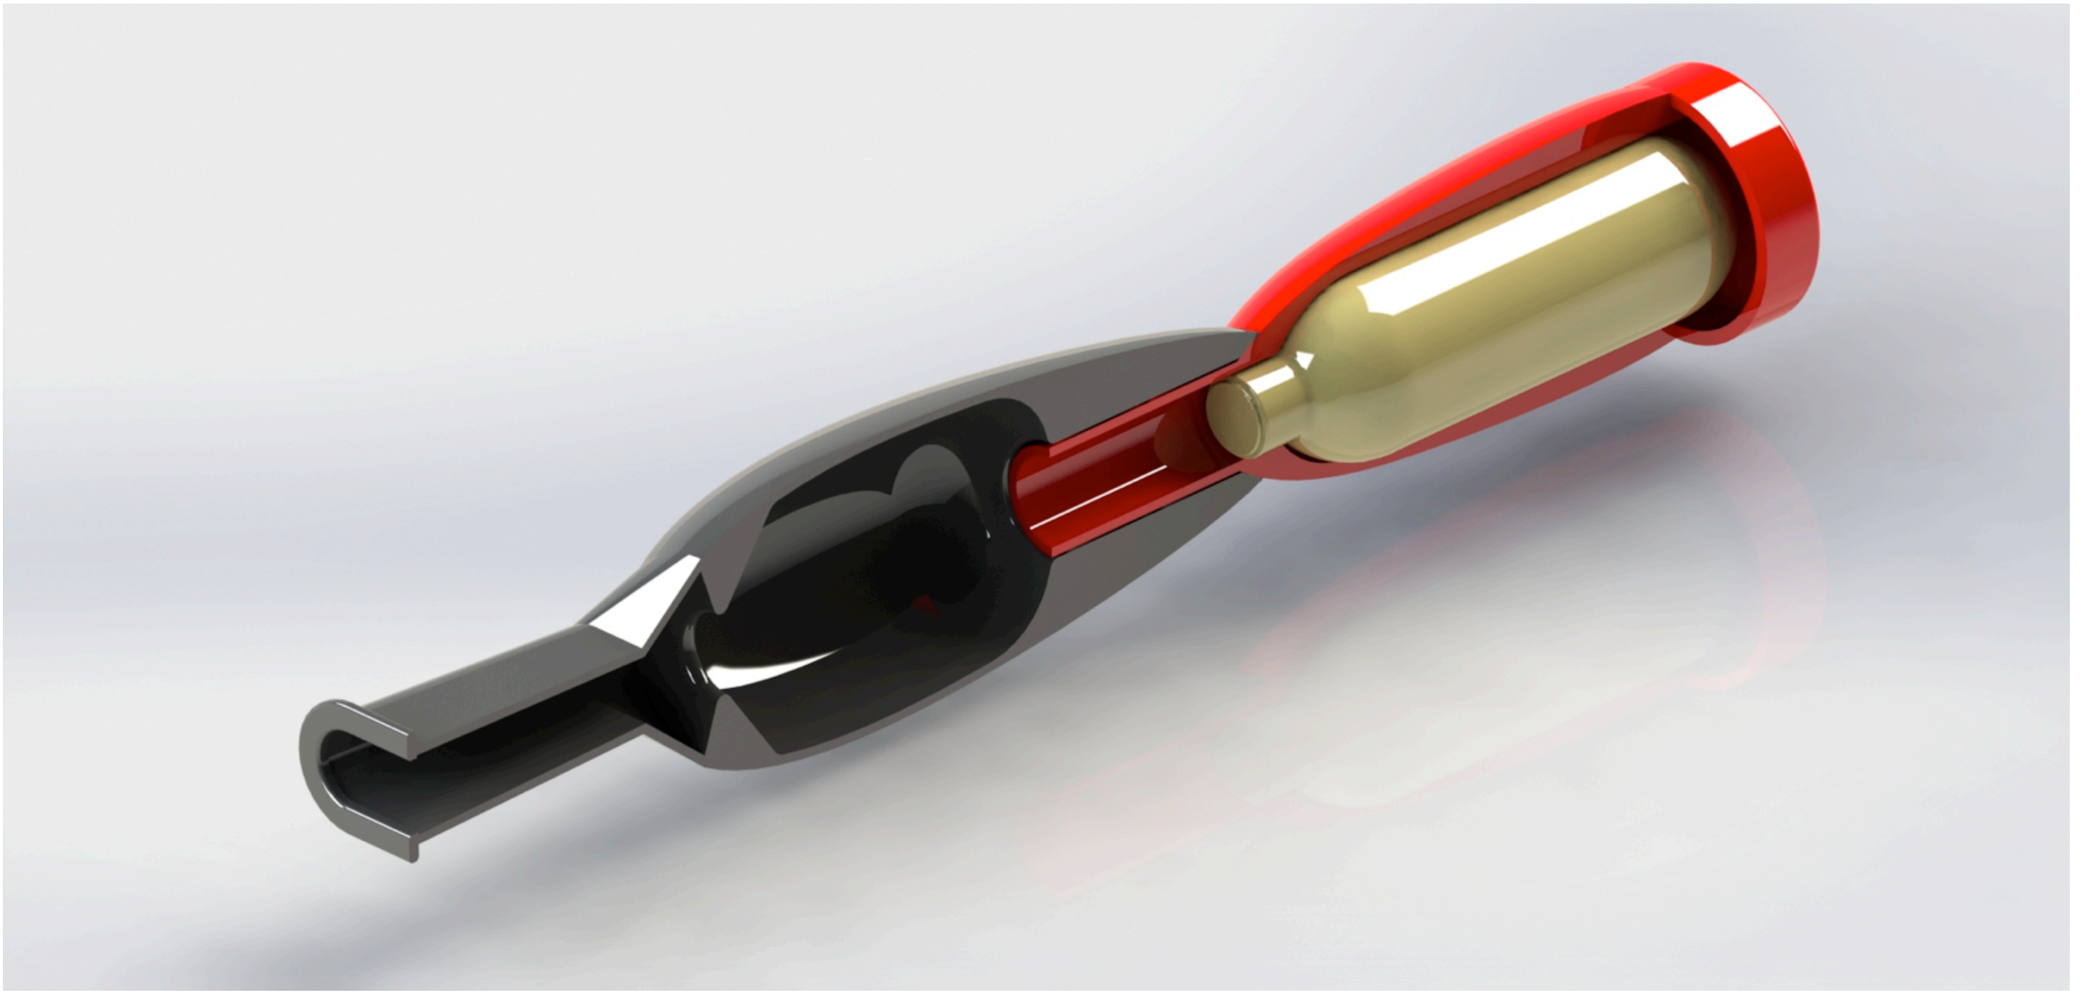

Novel Portable Delivery Device

S-1226™: Novel Delivery Device

- Portable rescue device to treat severe bronchoconstriction as a first line emergency treatment following an acute asthma attack
- Expected to complement and enhance activity of other drugs, particularly when conventional drugs are ineffective

Components of the Device Prototype

- Separate storage of a compressed gas cylinder and a drug-liquid ampoule within the device body
- On activation, compressed drug-gas is released through the liquid reservoir where PFOB vapor evaporates through an intermediate aerosol stage
- The combined gas and vapor mixture is then delivered to a tidally breathing patient at a controlled dosing rate and multi-minute treatment time

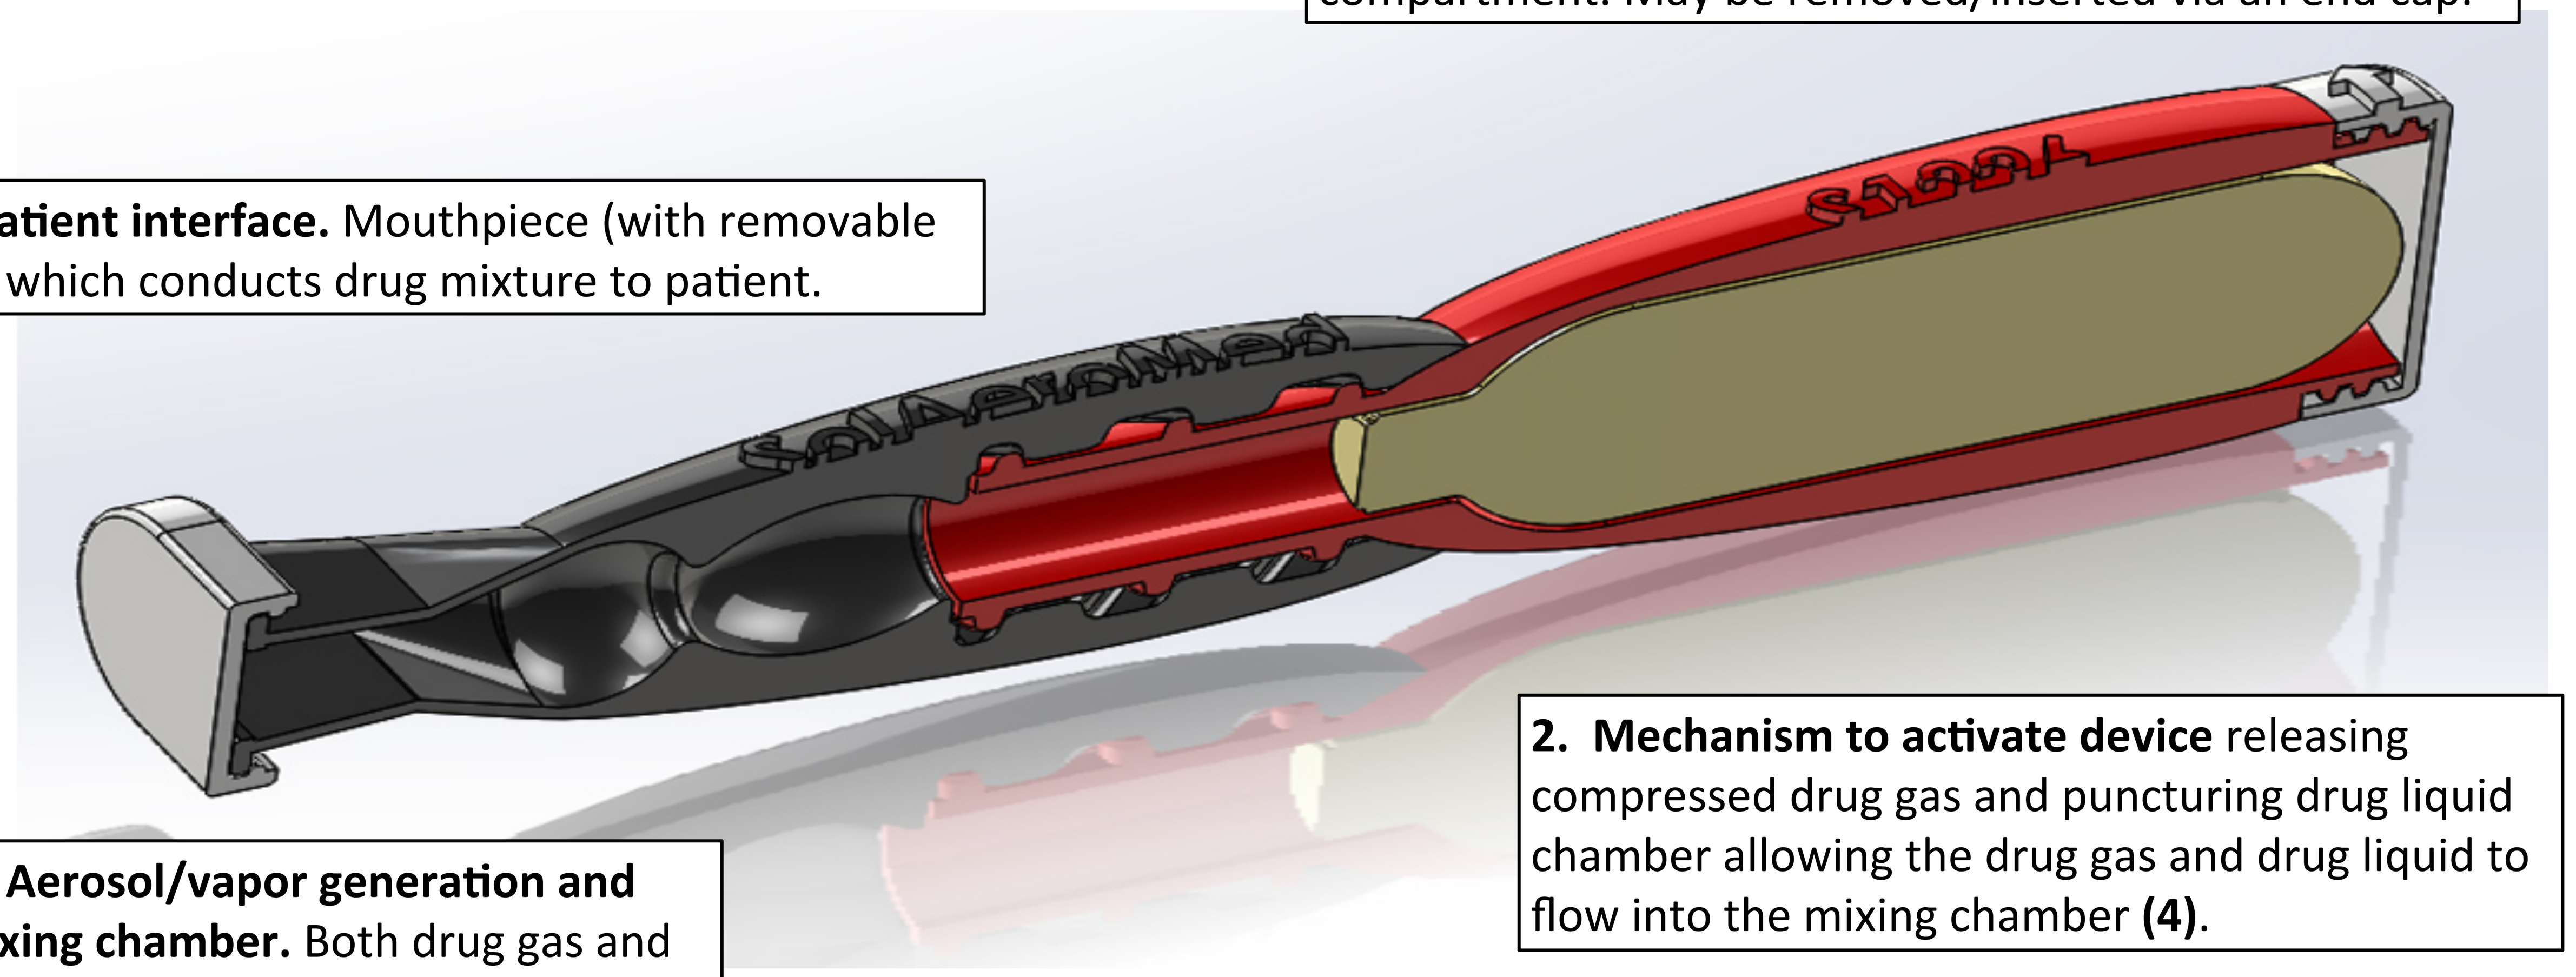
SolAeroMed
S1226

1. **Drug gas cartridge.** Resides within the terminal compartment. May be removed/inserted via an end cap.

2. **Mechanism to activate device** releasing compressed drug gas and puncturing drug liquid chamber allowing the drug gas and drug liquid to flow into the mixing chamber **(4)**.

3. **Drug liquid holding chamber** capable of storing liquid in a sealed environment until device is activated.

4. **Aerosol/vapor generation and mixing chamber.** Both drug gas and drug liquid enter allowing generation of liquid vapor and aerosol.

5. **Patient interface.** Mouthpiece (with removable cap) which conducts drug mixture to patient.

Summary and Future Directions

- **S-1226TM**:
 - Novel therapeutic bronchodilator that combines a potent bronchodilator (CO_2) with a synthetic lung surfactant (PFOB)
 - **CO_2**
 - Causes temporary rapid airway dilation via epithelial receptors and vagal C-fibers
 - Rapidly penetrates closed airways
 - **PFOB**
 - Provides some airway dilation and interacts with natural lung surfactant
 - Lubricates mucous plugs to enhance mucous clearance
 - First-in-class product; pharmacological and biophysical unique mechanism of action
 - May be developed to treat exacerbations of COPD and CF
- Novel portable delivery device to be used by patients as an emergency rescue technique
 - Platform technology for delivery of other drugs in the future
- **Clinical Trials:**
 - Q4 2015 – Completion of Phase IIa proof of concept clinical trial
 - Q3 2016 – Dose-ranging Phase II clinical trial
 - Q4 2016 – Repeat dosing Phase II clinical trial
 - Q4 2016 – Non-inferiority clinical trial for S-1226TM portable delivery device
- SolAeroMed Inc. is actively exploring licensing and partnering opportunities for our respiratory drug and delivery device technologies

For further information, contact:
Dr. John Dennis, CEO
www.solaeromed.com

* European Respiratory Society Conference, Amsterdam Sept 2015. Abstract 852023.