Biophysical Bronchodilator: A new class of respiratory drug*

John Dennis[1], Richard Leigh[2], Tamer El Mays[1], Cora Pieron[1], Gurkeet Lalli[1], Andrea Chiu[1], Morenike Fadayomi[1], Gareth Lewis[1], Francis Green[1,2]

[1]SolAeroMed Inc, Calgary, Alberta, Canada, T2L 2K8

[2]Respiratory Medicine, University of Calgary and Alberta Health Services, Calgary, Alberta, Canada

Abstract

- We have discovered a new short acting bronchodilator which has a different mechanism of action than current bronchodilators. The drug, **S-1226™,** is a novel formulation of the potent bronchodilator carbon dioxide (4-12% CO_2 enriched air) combined with a synthetic lung surfactant, perfluoroctylbromide (PFOB), delivered as a nebulized mixture of vapour and aerosol to the lungs.
- **S-1226™**:
 - Effective: Treats methacholine and allergen induced bronchoconstriction in sheep and rat models of asthma
 - Safe: Phase I clinical trial showed minimal adverse effects at all dosing levels (4%, 8%, 12% CO_2)
 - Currently in Phase II clinical trial – allergen induced asthmatic subjects
- **S-1226™** has a unique biophysical mechanism of action
 - Neither adrenergic nor cholinergic
 - CO_2 causes temporary rapid airway dilation via epithelial receptors and vagal C-fibers
 - PFOB provides some airway dilation and interacts with natural lung surfactant
 - Synergistic effect with combination of PFOB and CO_2
 - Very fast dilation expected to last 30-60 minutes
 - The ability of the gaseous component to rapidly penetrate closed airways
 - The ability to lubricate mucous plugs enhancing mucous clearance
- **S-1226™** is being developed by academic co-founders through the Canadian based biotech start-up SolAeroMed Inc.

Medical Need for a New Asthma Treatment

- Asthma:
 - Affects 300 million individuals worldwide; 400 million expected by 2025
 - 250,000 asthma deaths every year worldwide
- Existing treatments:
 - Short-acting β_2-agonists are the front-line emergency treatment
 - Salbutamol is the only short-acting bronchodilator available following an asthma attack
 - One-fifth to one-third of patients do **not** respond to this therapy
- When treatment is ineffective:
 - **3.7 million** asthma-related emergency department visits per year worldwide
 - Patients are treated with higher doses of same drug (Salbutamol) that failed
 - No other alternative exists
 - $5.3 billion/year in hospital costs
- Major challenge for treating acute asthma is to open occluded airways rapidly enough to re-establish ventilation and allow delivery of O_2 and conventional medication to airways
- A new treatment is required that has pharmacological properties (bronchodilation) and biophysical properties (mucous plug penetration and clearance) for a more rapid and effective therapy

S-1226™: Novel Bronchodilator

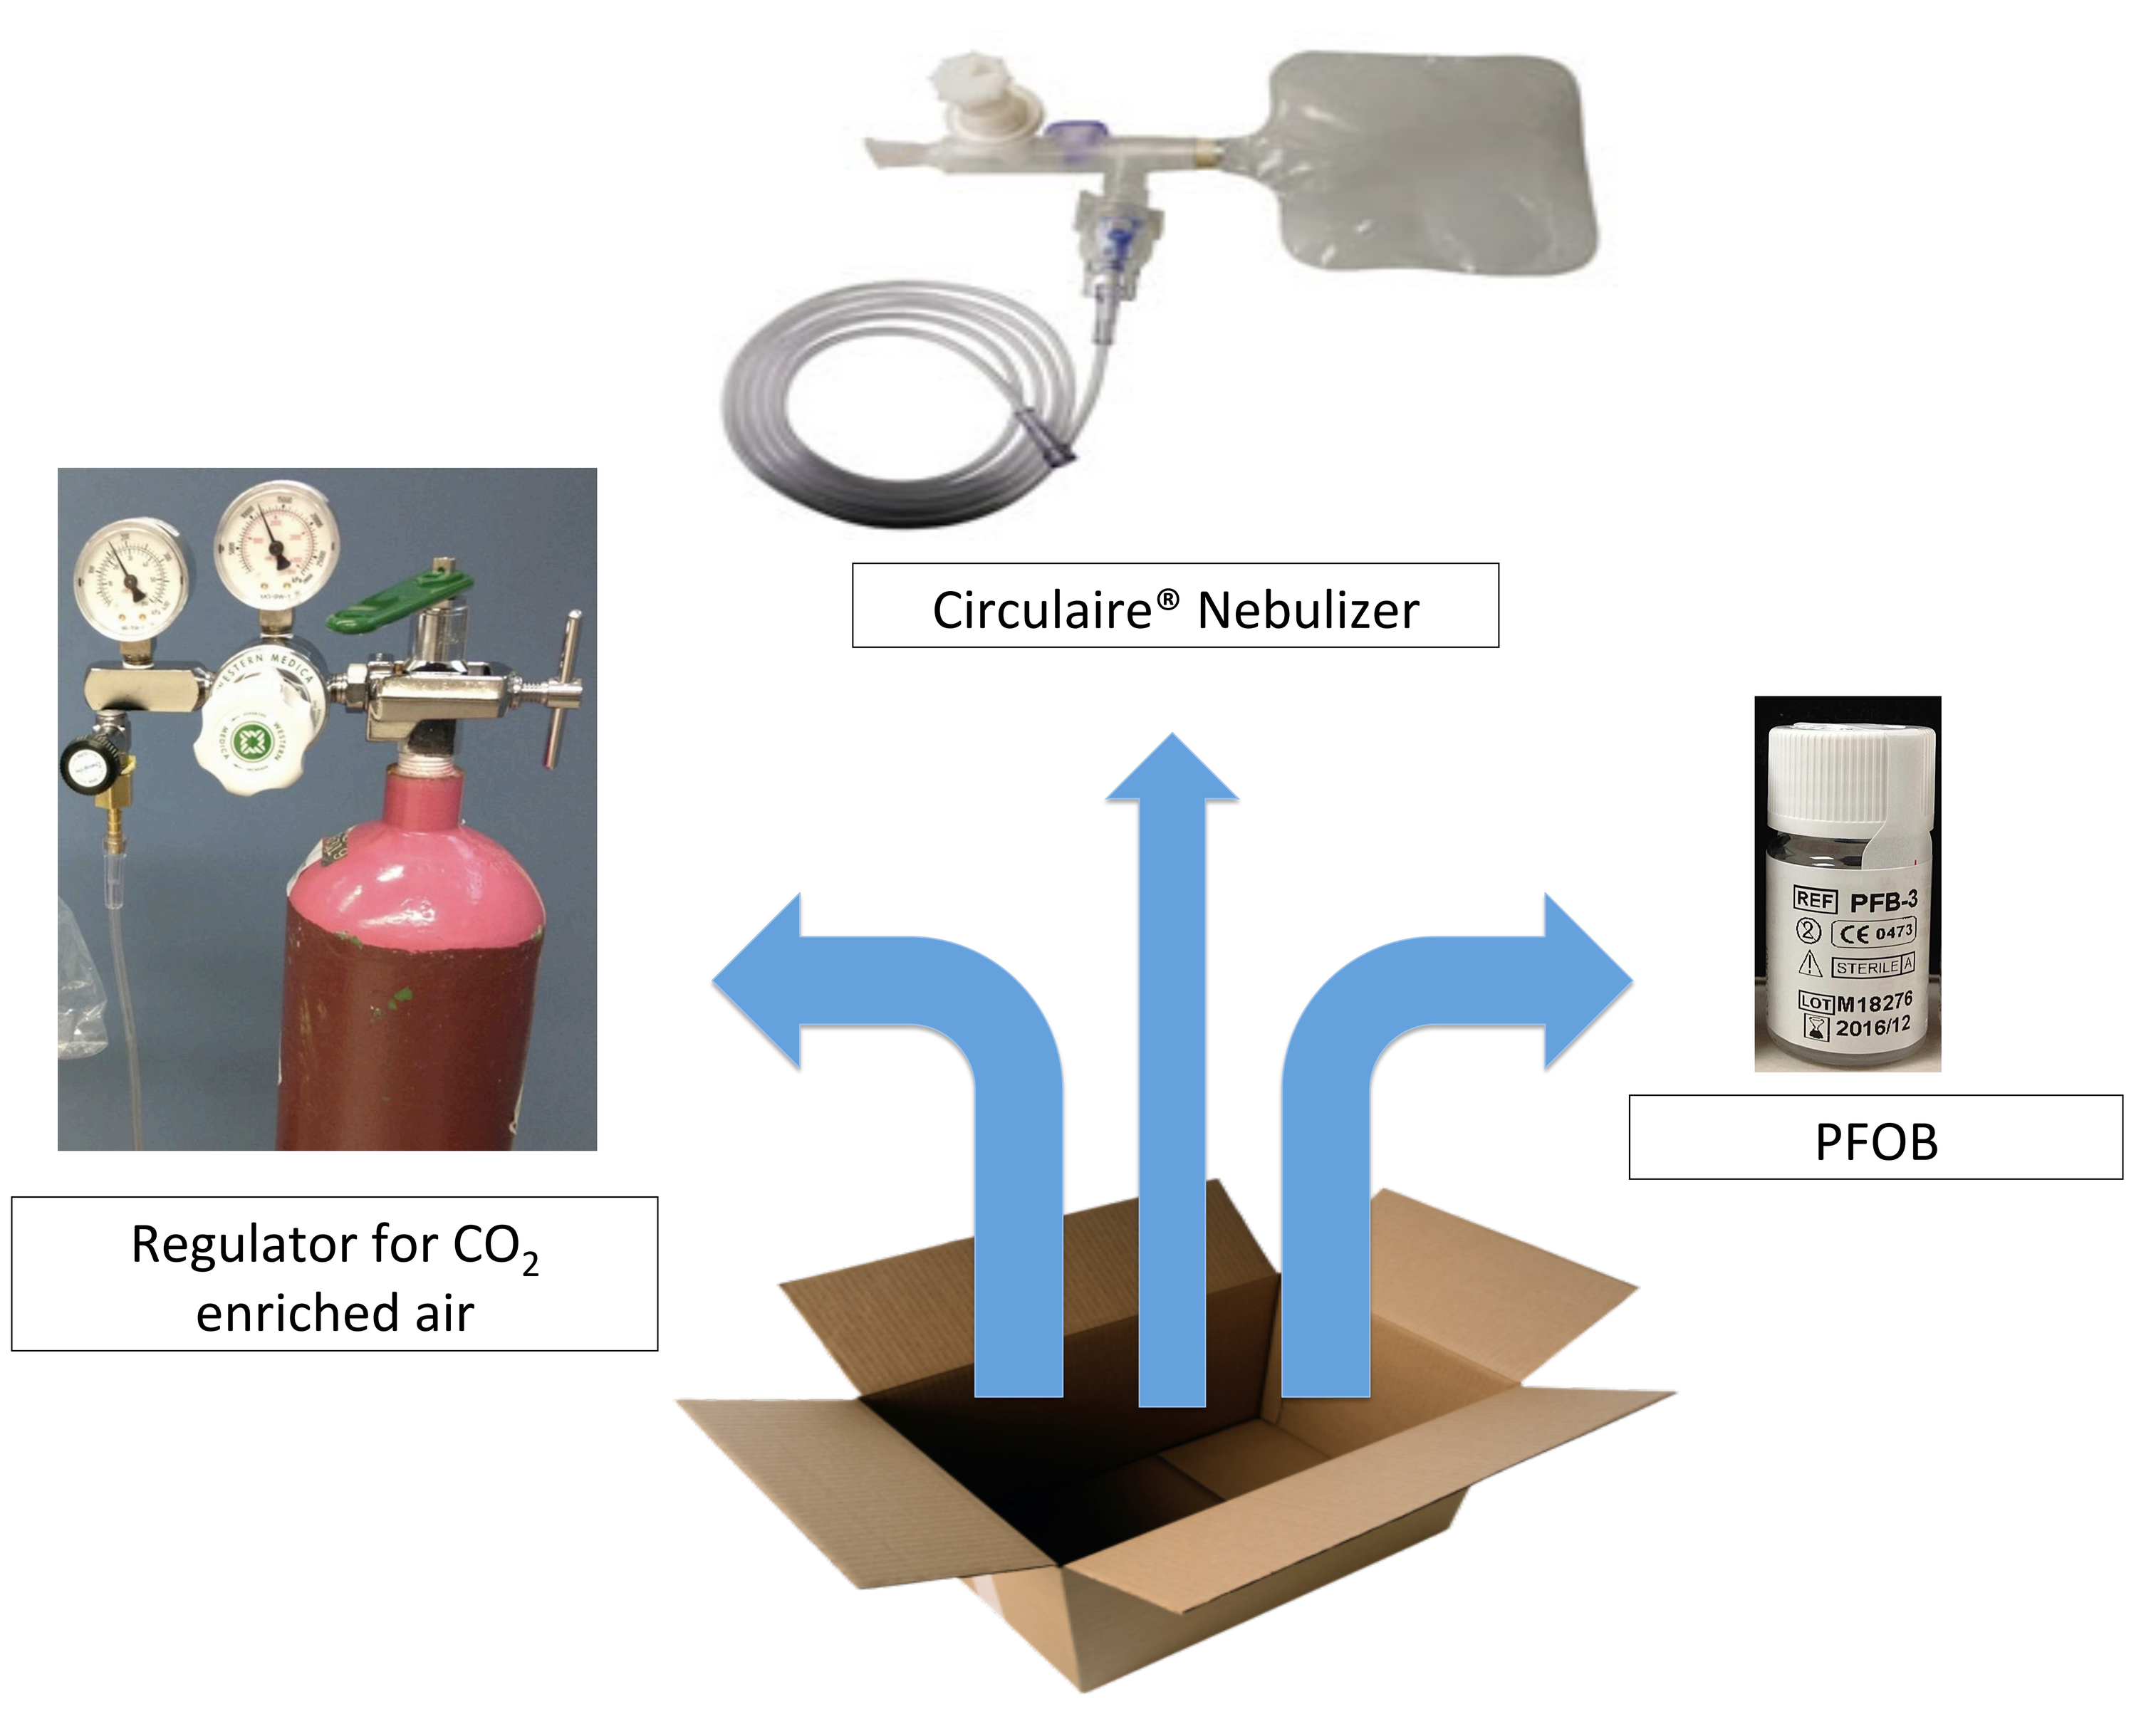
Circulaire® Nebulizer
Regulator for CO_2 enriched air
PFOB
REF PFB-3
LOT M18276
2016/12

- S-1226™ consists of 3mL PFOB synthetic surfactant nebulized with CO_2-enriched air (4-12% CO_2, 20% O_2, with balanced N_2)
 - Delivered as an aerosol/vapour/gas mixture
 - Short-acting bronchodilator with a novel mechanism of action
 - Both CO_2 and PFOB are well-described and safe
 - Both CO_2 and PFOB have previously been approved by FDA, EMA, HC

Pre-Clinical Studies

- S-1226™ has a fast and long-lasting bronchodilator effect in sheep model

Figure 1

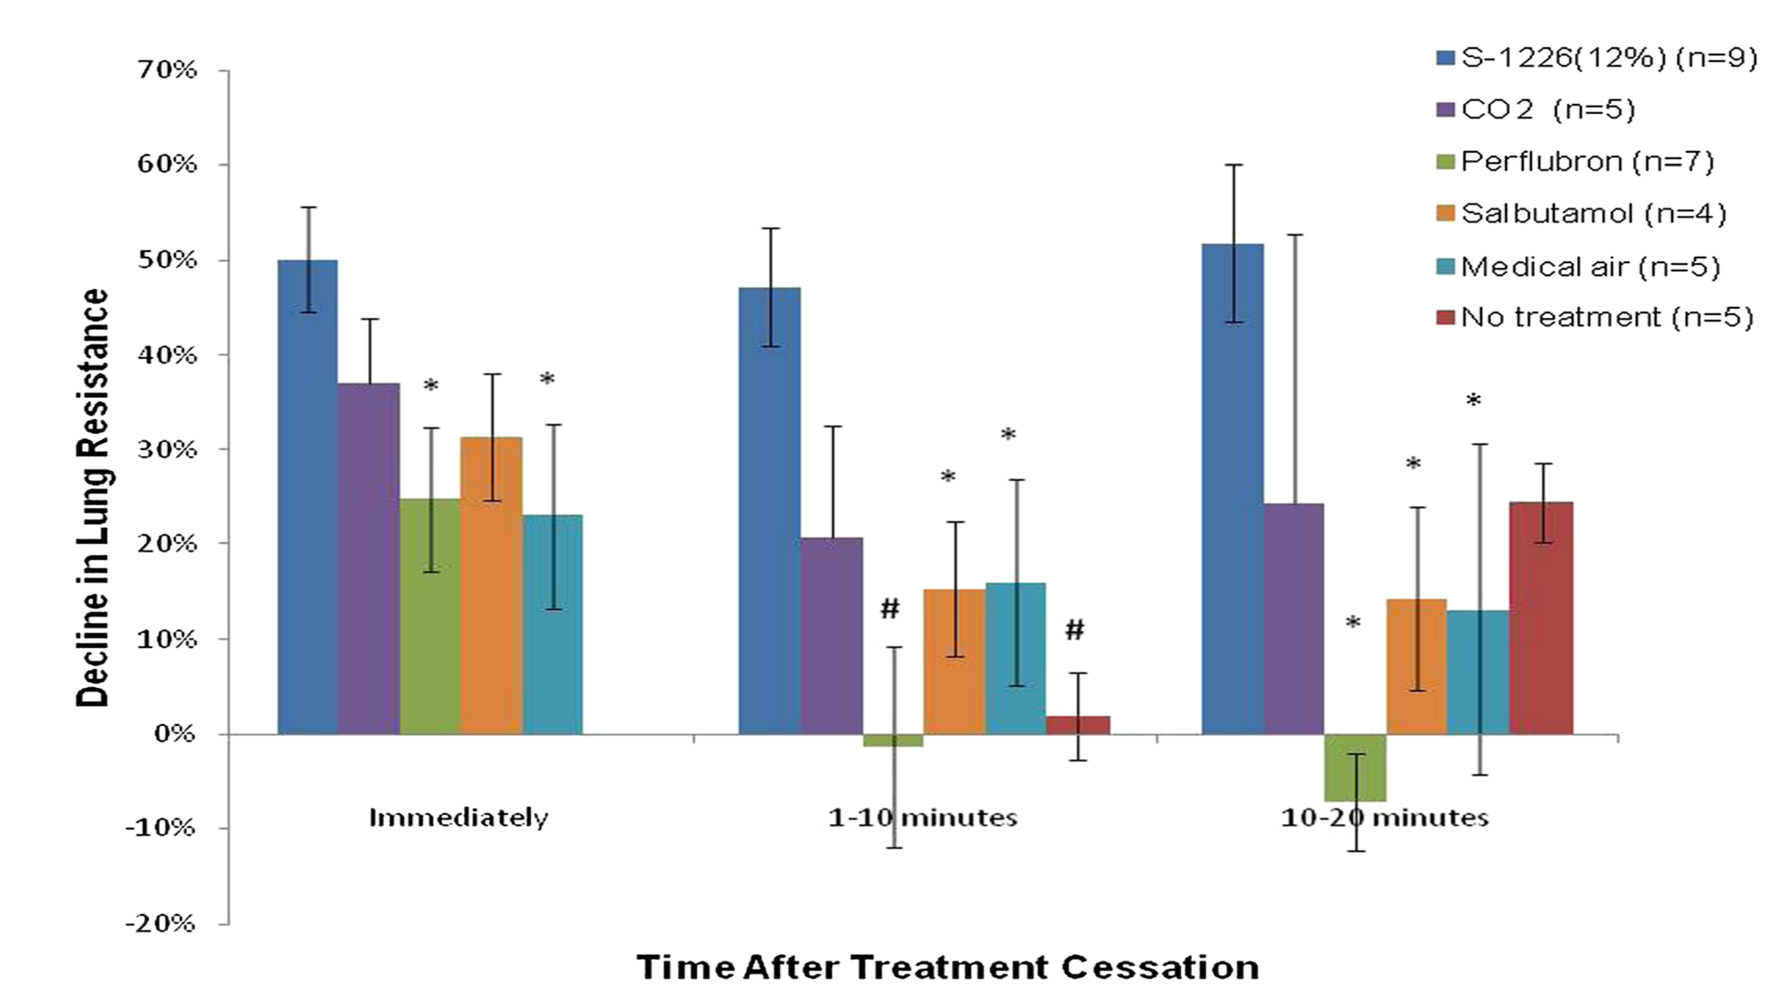
S-1226(12%) (n=9)
CO2 (n=5)
Perflubron (n=7)
Salbutamol (n=4)
Medical air (n=5)
No treatment (n=5)
Decline in Lung Resistance
70%
60%
50%
40%
30%
20%
10%
0%
-10%
-20%
Immediately
1-10 minutes
10-20 minutes
Time After Treatment Cessation

Figure 2

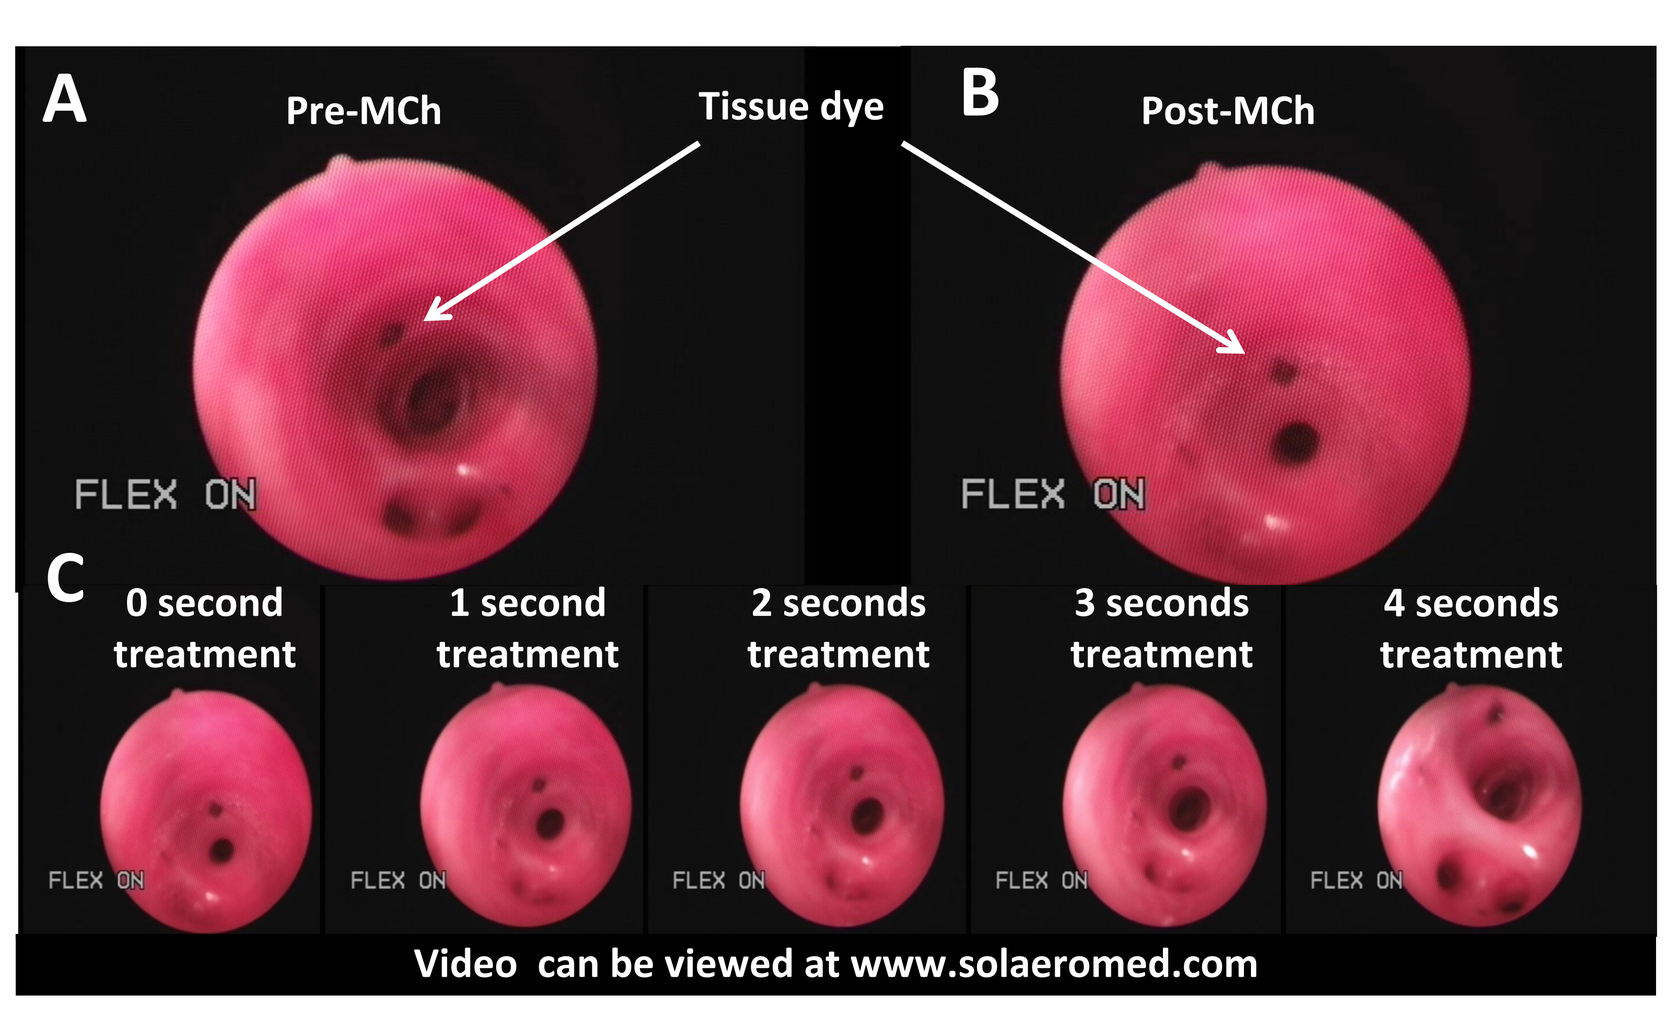
A
Pre-MCh
Tissue dye
B
Post-MCh
FLEX ON
C
0 second treatment
1 second treatment
2 seconds treatment
3 seconds treatment
4 seconds treatment
Video can be viewed at www.solaeromed.com

Figure 1: Decline in lung resistance following treatment in house dust mite (HDM) challenged sheep. Treatment with S-1226™ showed significant and sustained declines in lung resistance compared to medical air or no treatment. S-1226™ showed a significantly greater decline in lung resistance compared to salbutamol. P-values show significant difference from S-1226™; *p <0.05, # p ≤ 0.01.
Figure 2: Bronchoscopic still images of sheep airways before (A) and after (B) methacholine (MCh) challenge and seconds after initiating treatment with nebulised S-1226™. (C) Complete airway opening is seen 4 seconds after the start of S-1226™ treatment.

Clinical Studies

- **Phase I completed 2014 (publication pending)**
 - N=36 healthy patients, GLP compliant
 - Inhaled 2 minute treatment of S-1226™, dose increase from 4% CO_2, 8% CO_2 & 12% CO_2
 - No safety concerns reported
- **Phase IIa Proof of Concept (in progress; completion expected Q4 2015)**
 - Allergen induced asthma, n=12 subjects
 - Double blind crossover, GLP compliant
 - Placebo (saline & air) Vs. S1226 (8% CO_2 & PFOB)
 - Aim to evaluate safety and efficacy
- **Near Term Catalysts (2015-2016)**

Event	Description	Timeline
Phase 2 Read-out Proof of Concept	Proof of Concept, n=12 single center, double blind cross over in Allergen induced asthmatics.	Q3 2015
Phase II Dose Ranging	Dose escalation & multiple consecutive dosing in allergen induced asthmatics	Q3 2016
Phase II Repeat Dosing	Multiple consecutive dosings in allergen induced asthmatics	Q4 2016
Phase II Non-Inferiority	Portable delivery device Outpatient bioequivalence	Q4 2016

Summary and Future Directions

- **S-1226™:**
 - Rapidly dilates airways (*in vivo*) and enhances mucous plug clearance (*in vitro*) using a new mechanism of action thus rapidly opening closed airways and enhancing delivery of standard bronchodilation therapies
 - First-in-class product; pharmacological and biophysical mechanism of action
 - May be developed to treat COPD and CF exacerbations in the future

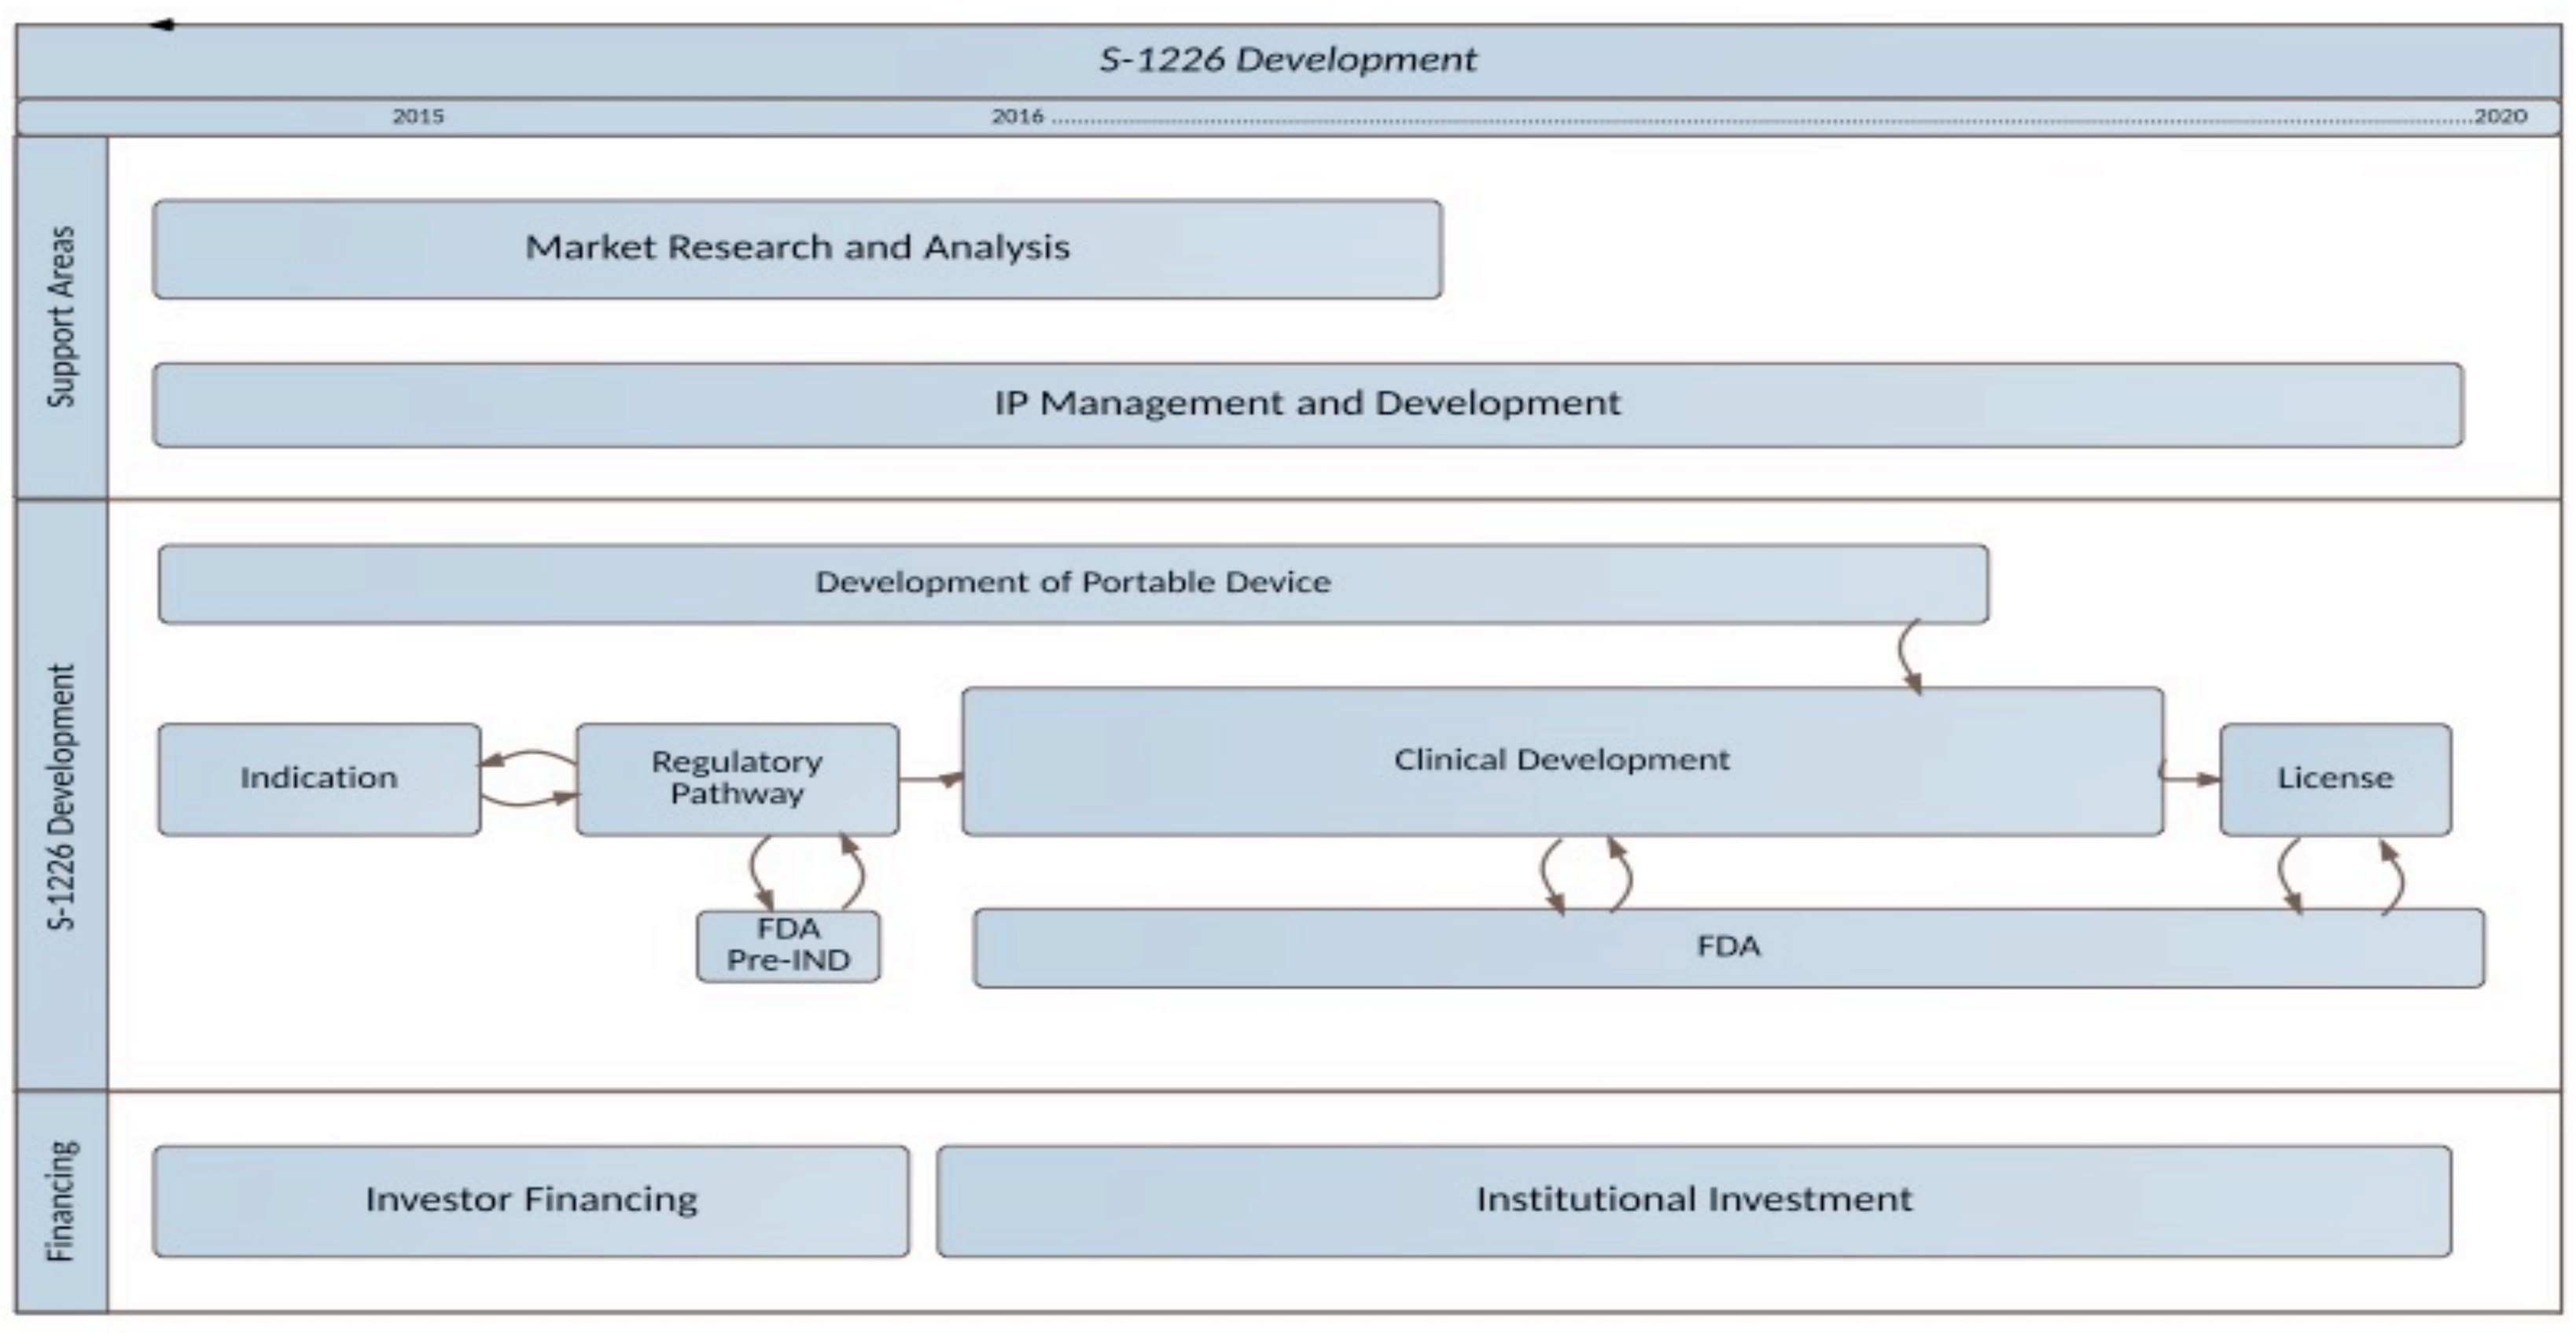
S-1226 Development
2015
2016
2020
Support Areas
Market Research and Analysis
IP Management and Development
S-1226 Development
Development of Portable Device
Indication
Regulatory Pathway
Clinical Development
License
FDA Pre-IND
FDA
Financing
Investor Financing
Institutional Investment

- Development of a personal rescue device to be used by patients as a first line emergency treatment following an acute asthma attack
- SolAeroMed Inc. is actively exploring licensing and partnering opportunities for our respiratory drugs and delivery device technologies

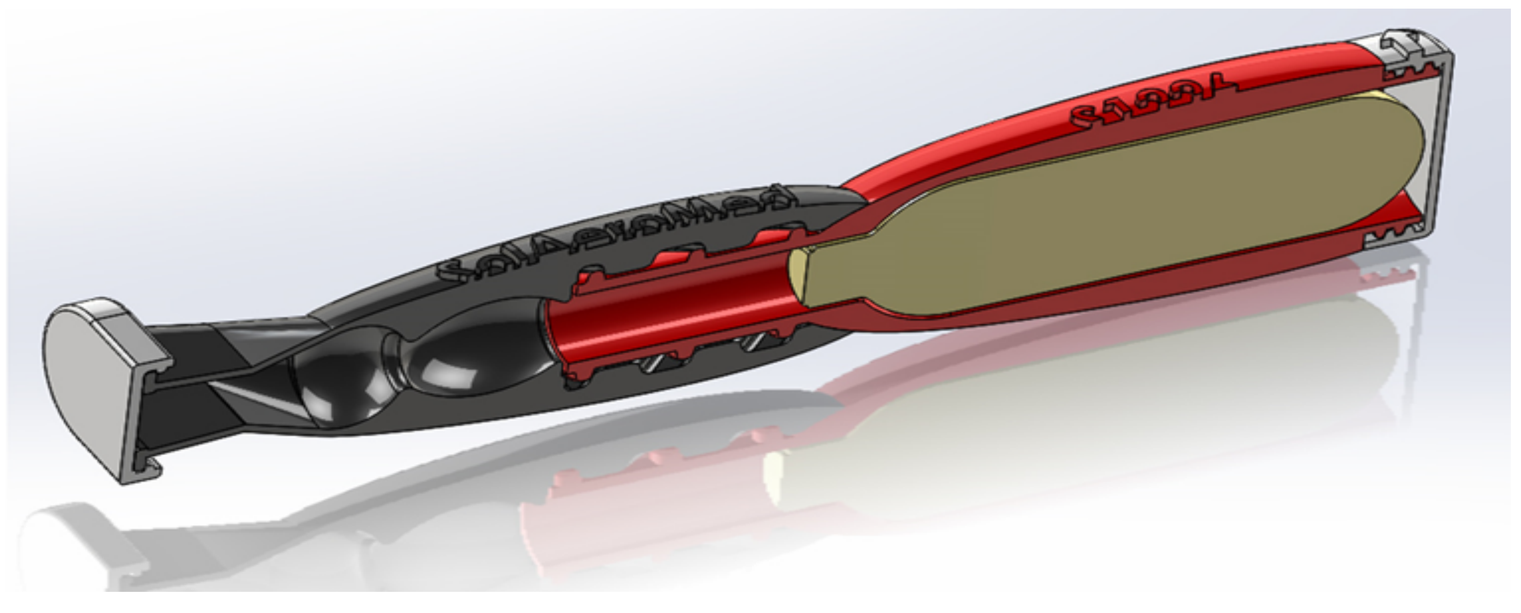

For more information , contact:
Dr. John Dennis, CEO
www.solaeromed.com

* European Respiratory Society Conference, Amsterdam Sept 2015. Abstract #854490.